XPO1 Inhibition Suppresses the Growth of Pancreatic Neuroendocrine Tumors

Md. Hafiz Uddin[1], Sahar Bannoura[1], Amro Aboukameel[1], Mohammed Najeeb Al-Hallak[1], Yosef Landesman[2], Ramzi M. Mohammad[1], Philip A. Philip[1], Bassel El-Rayes[3], Asfar S. Azmi[1]*

[1]Department of Oncology, Wayne State University School of Medicine, Detroit MI 48201 USA; [2]Karyopharm Therapeutics Inc., Newton, MA 02459, USA; [3]Emory Winship Cancer Institute, Atlanta, Georgia.

Abstract

Pancreatic neuroendocrine tumors (PNETs) are rare islet cell tumors. Although slow growing in early stages, the overall survival rates of metastatic PNETs are dismally low at 25%. The main treatment option includes surgery followed by chemotherapy or targeted therapy. Unfortunately, advanced PNETs show minimal response to FDA approved therapies suggesting the urgent need for the identification of novel and effective treatments. In the present study, we have tested 1st and 2nd generation XPO1 inhibitors also known as selective inhibitor of nuclear export (SINE) on BON1 and QGP1 PNET tumor cells. The IC_{50s} for SINE compounds namely KPT-185, KPT-330 (selinexor/XPOVIO), KPT-8602 (eltanexor) were 26 nM, 283 nM, 1027 nM respectively for BON1 cells. Similar trends in the IC_{50s} were observed in QGP1 cells. Both the 1st and 2nd generation SINE, KPT-330 and KPT-8602 reduced the number and area of the colonies significantly. SINE compounds were able to induce apoptosis at pharmacologically relevant concentrations. Western blot analysis also revealed significant induction of PARP cleavage by SINE. The PNET marker Chromogranin A was found to be reduced in the SINE treated cells in IF assay. XPO1 inhibitors also suppress pmTOR and pP70S6K along with mTORC2 pathway molecule RICTOR in QGP1 cells. Taken together, this is the first study to reveal the therapeutic potential of novel XPO1 targeted agents for the treatment of PNETs. The *in vivo* evaluations of SINE compounds in xenograft models are underway.

Introduction

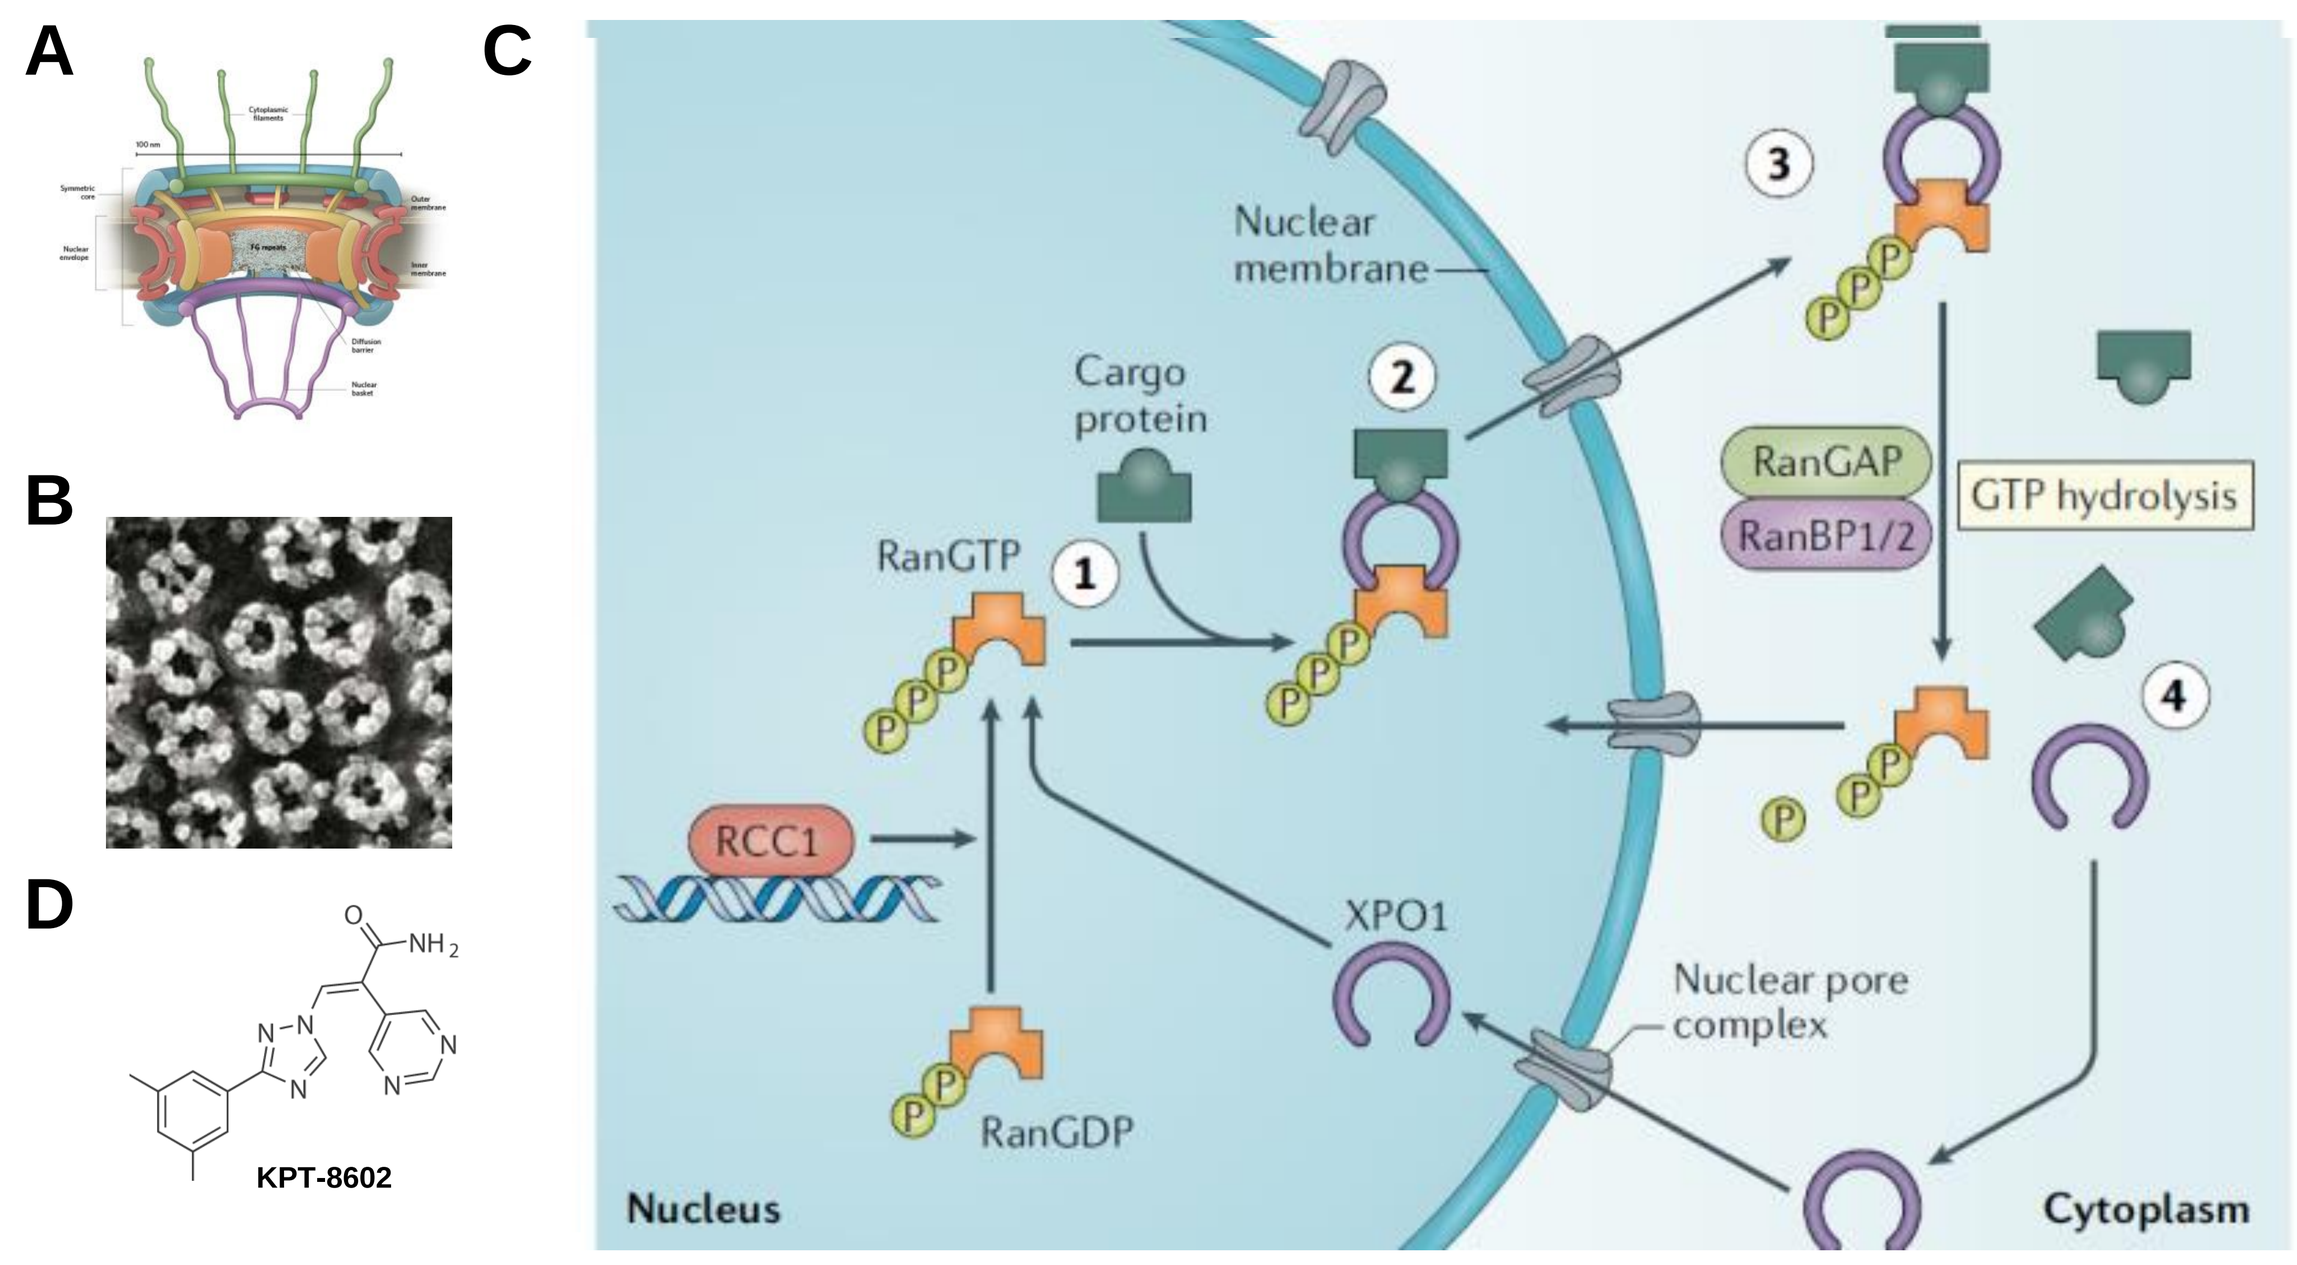
A
B
C
D
KPT-8602
Nuclear membrane
Cargo protein
RanGTP
RanGAP
RanBP1/2
GTP hydrolysis
RCC1
XPO1
Nuclear pore complex
RanGDP
Nucleus
Cytoplasm

Figure 1. Schematic diagrams. A. Diagram of nuclear pore complex (NPC). B. Electron microphotograph of NPC. C. Mechanism of nuclear export: 1) Exportin-1 (CRM1/XPO1) hydrophobic groove binds to the leucine rich nuclear export signal (NES) domain of the cargo proteins. RanGTP and protein cargos bind to exportin-1 forming stable ternary and activate it by 3D conformational change. 2) Activated complex binds to NPC, a large supramolecular complex composed of more than 30 different proteins, the nucleoporins. 3) XPO1-complex passes through NPC and enters cytoplasm. 4) In the cytoplasm, the ternary exportin-1-cargo-RanGTP complexes are dissociated. Excessive nuclear export by XPO1 causes mislocalization dependent inactivation of tumor suppressors. Selective inhibitor of nuclear export (SINE) compounds (KPT-8602) bind to XPO1 and block nuclear export function. D. Structure of next generation SINE compound KPT-8602 (eltanexor).

Materials and Methods

- Growth inhibition was determined by MTT assay. Dose response non-linear curves were generated and IC_{50s} were calculated using GrpahPad Prism software.
- Colony forming ability was determined by clonogenic assay. No of colonies and area of the colonies were determined by NIH imageJ software.
- Apoptotic cell death was determined by flow cytometric analysis using annexin V and propidium iodide.
- Western blotting analysis was performed to detect protein levels.
- Expression of PNET marker Chromogranin A was determined using immunofluorescence (IF) technique.

Results

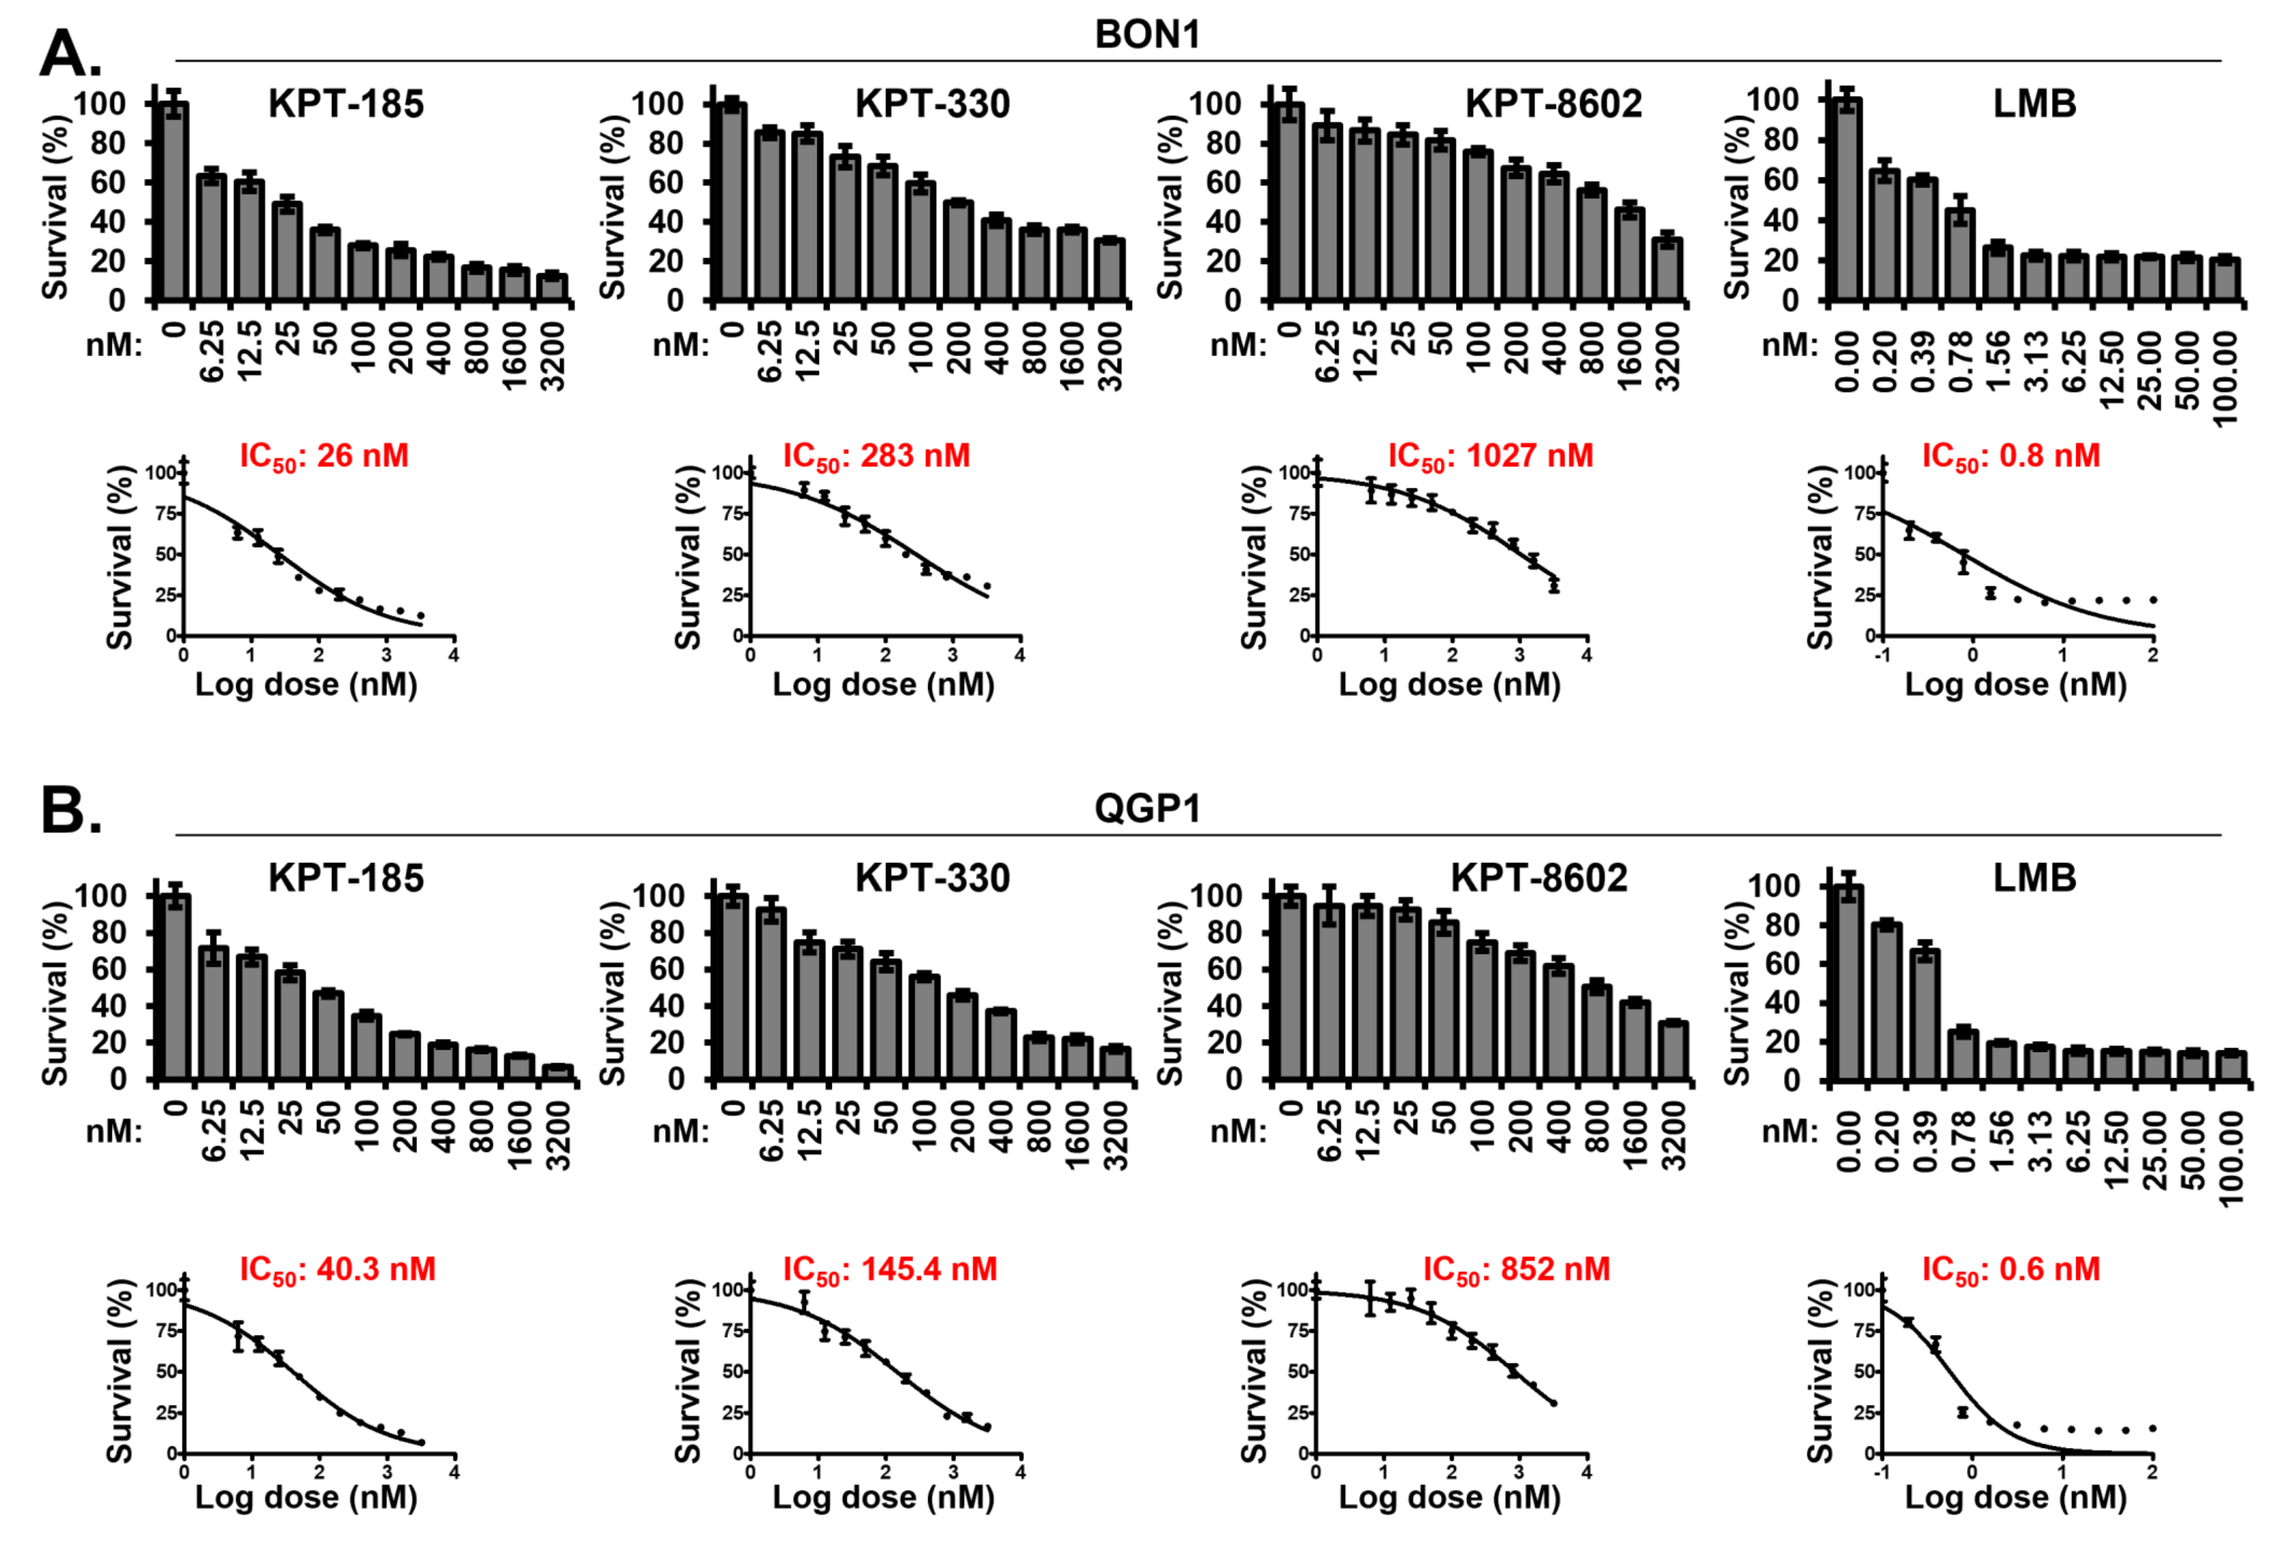
A.
BON1
KPT-185
KPT-330
KPT-8602
LMB
IC50: 26 nM
IC50: 283 nM
IC50: 1027 nM
IC50: 0.8 nM
B.
QGP1
IC50: 40.3 nM
IC50: 145.4 nM
IC50: 852 nM
IC50: 0.6 nM
Survival (%)
Log dose (nM)

Figure 2. Selective inhibitor of nuclear export (SINE) compounds (KPT-185, KPT-330 and KPT-8602) induced growth inhibition in pancreatic neuroendocrine tumors (PNET) cells. Leptomycin B (LMB) is the irreversible inhibitor of XPO1 and used as a positive control. A. BON1 cells. B. QGP1 cells. Cells were treated with varying doses of drugs for 72 hours. MTT assay was performed to determine the growth inhibition.

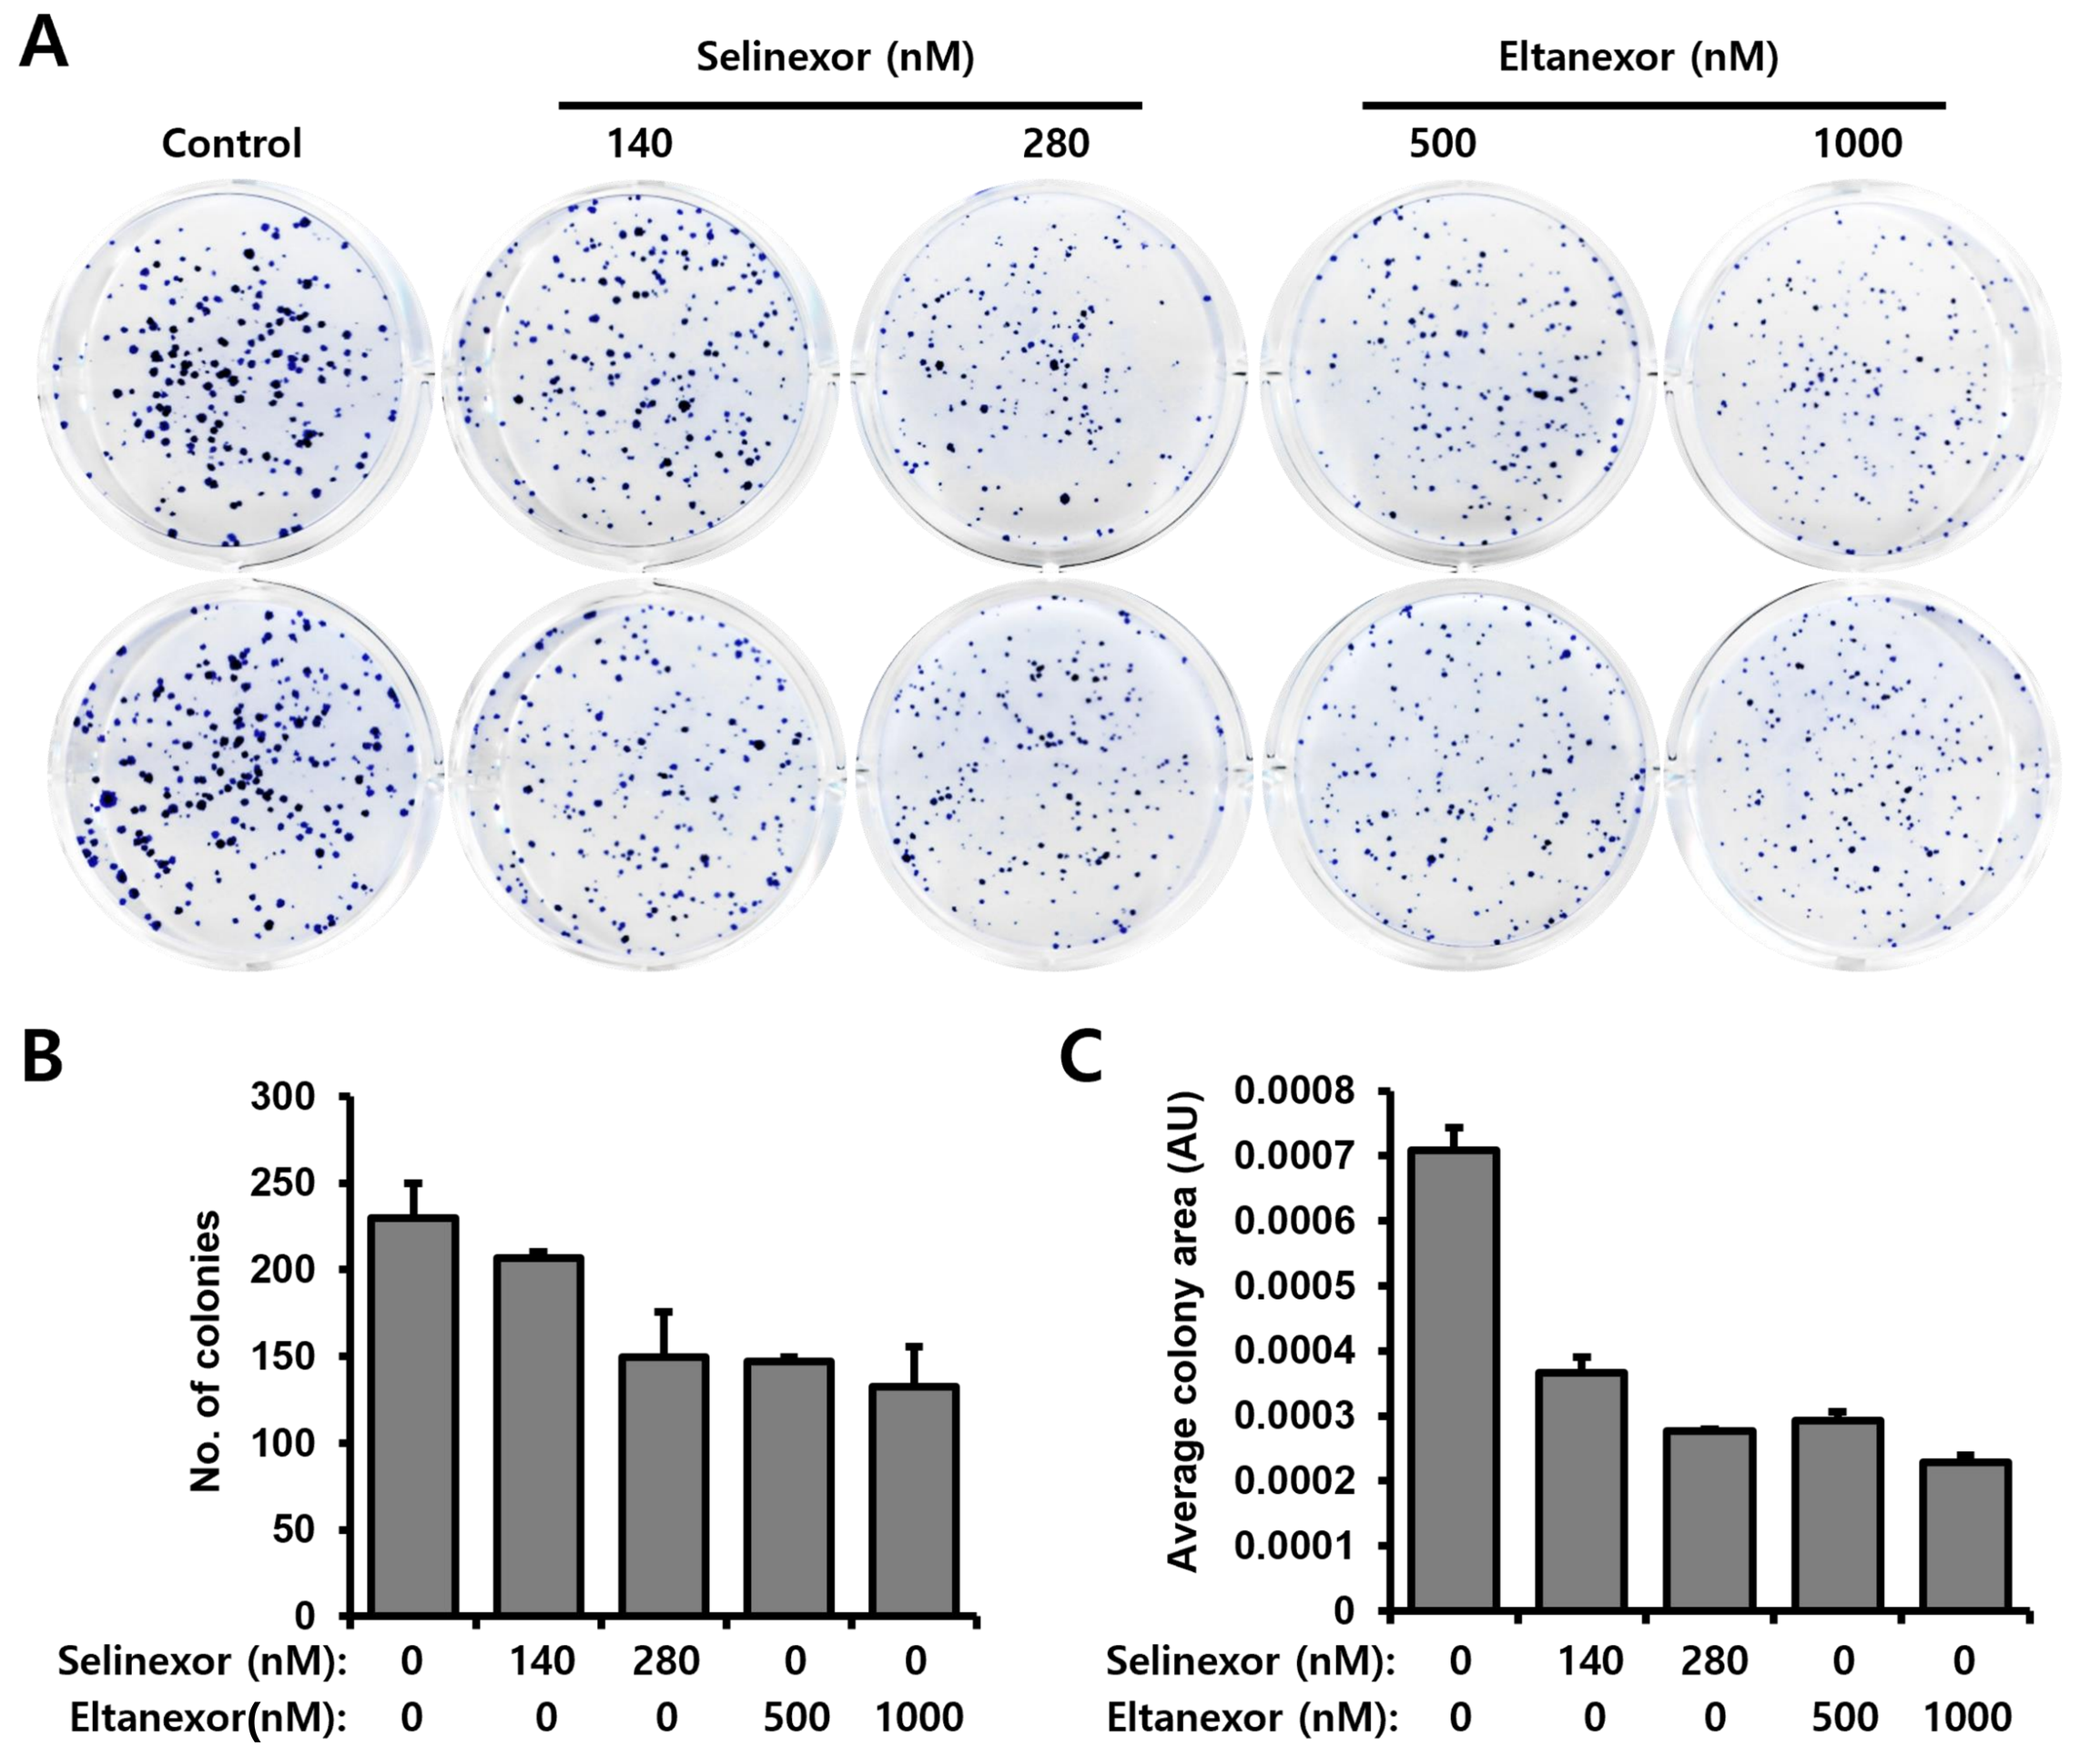
A
Selinexor (nM)
Eltanexor (nM)
Control
140
280
500
1000
B
No. of colonies
C
Average colony area (AU)
Selinexor (nM):
Eltanexor(nM):

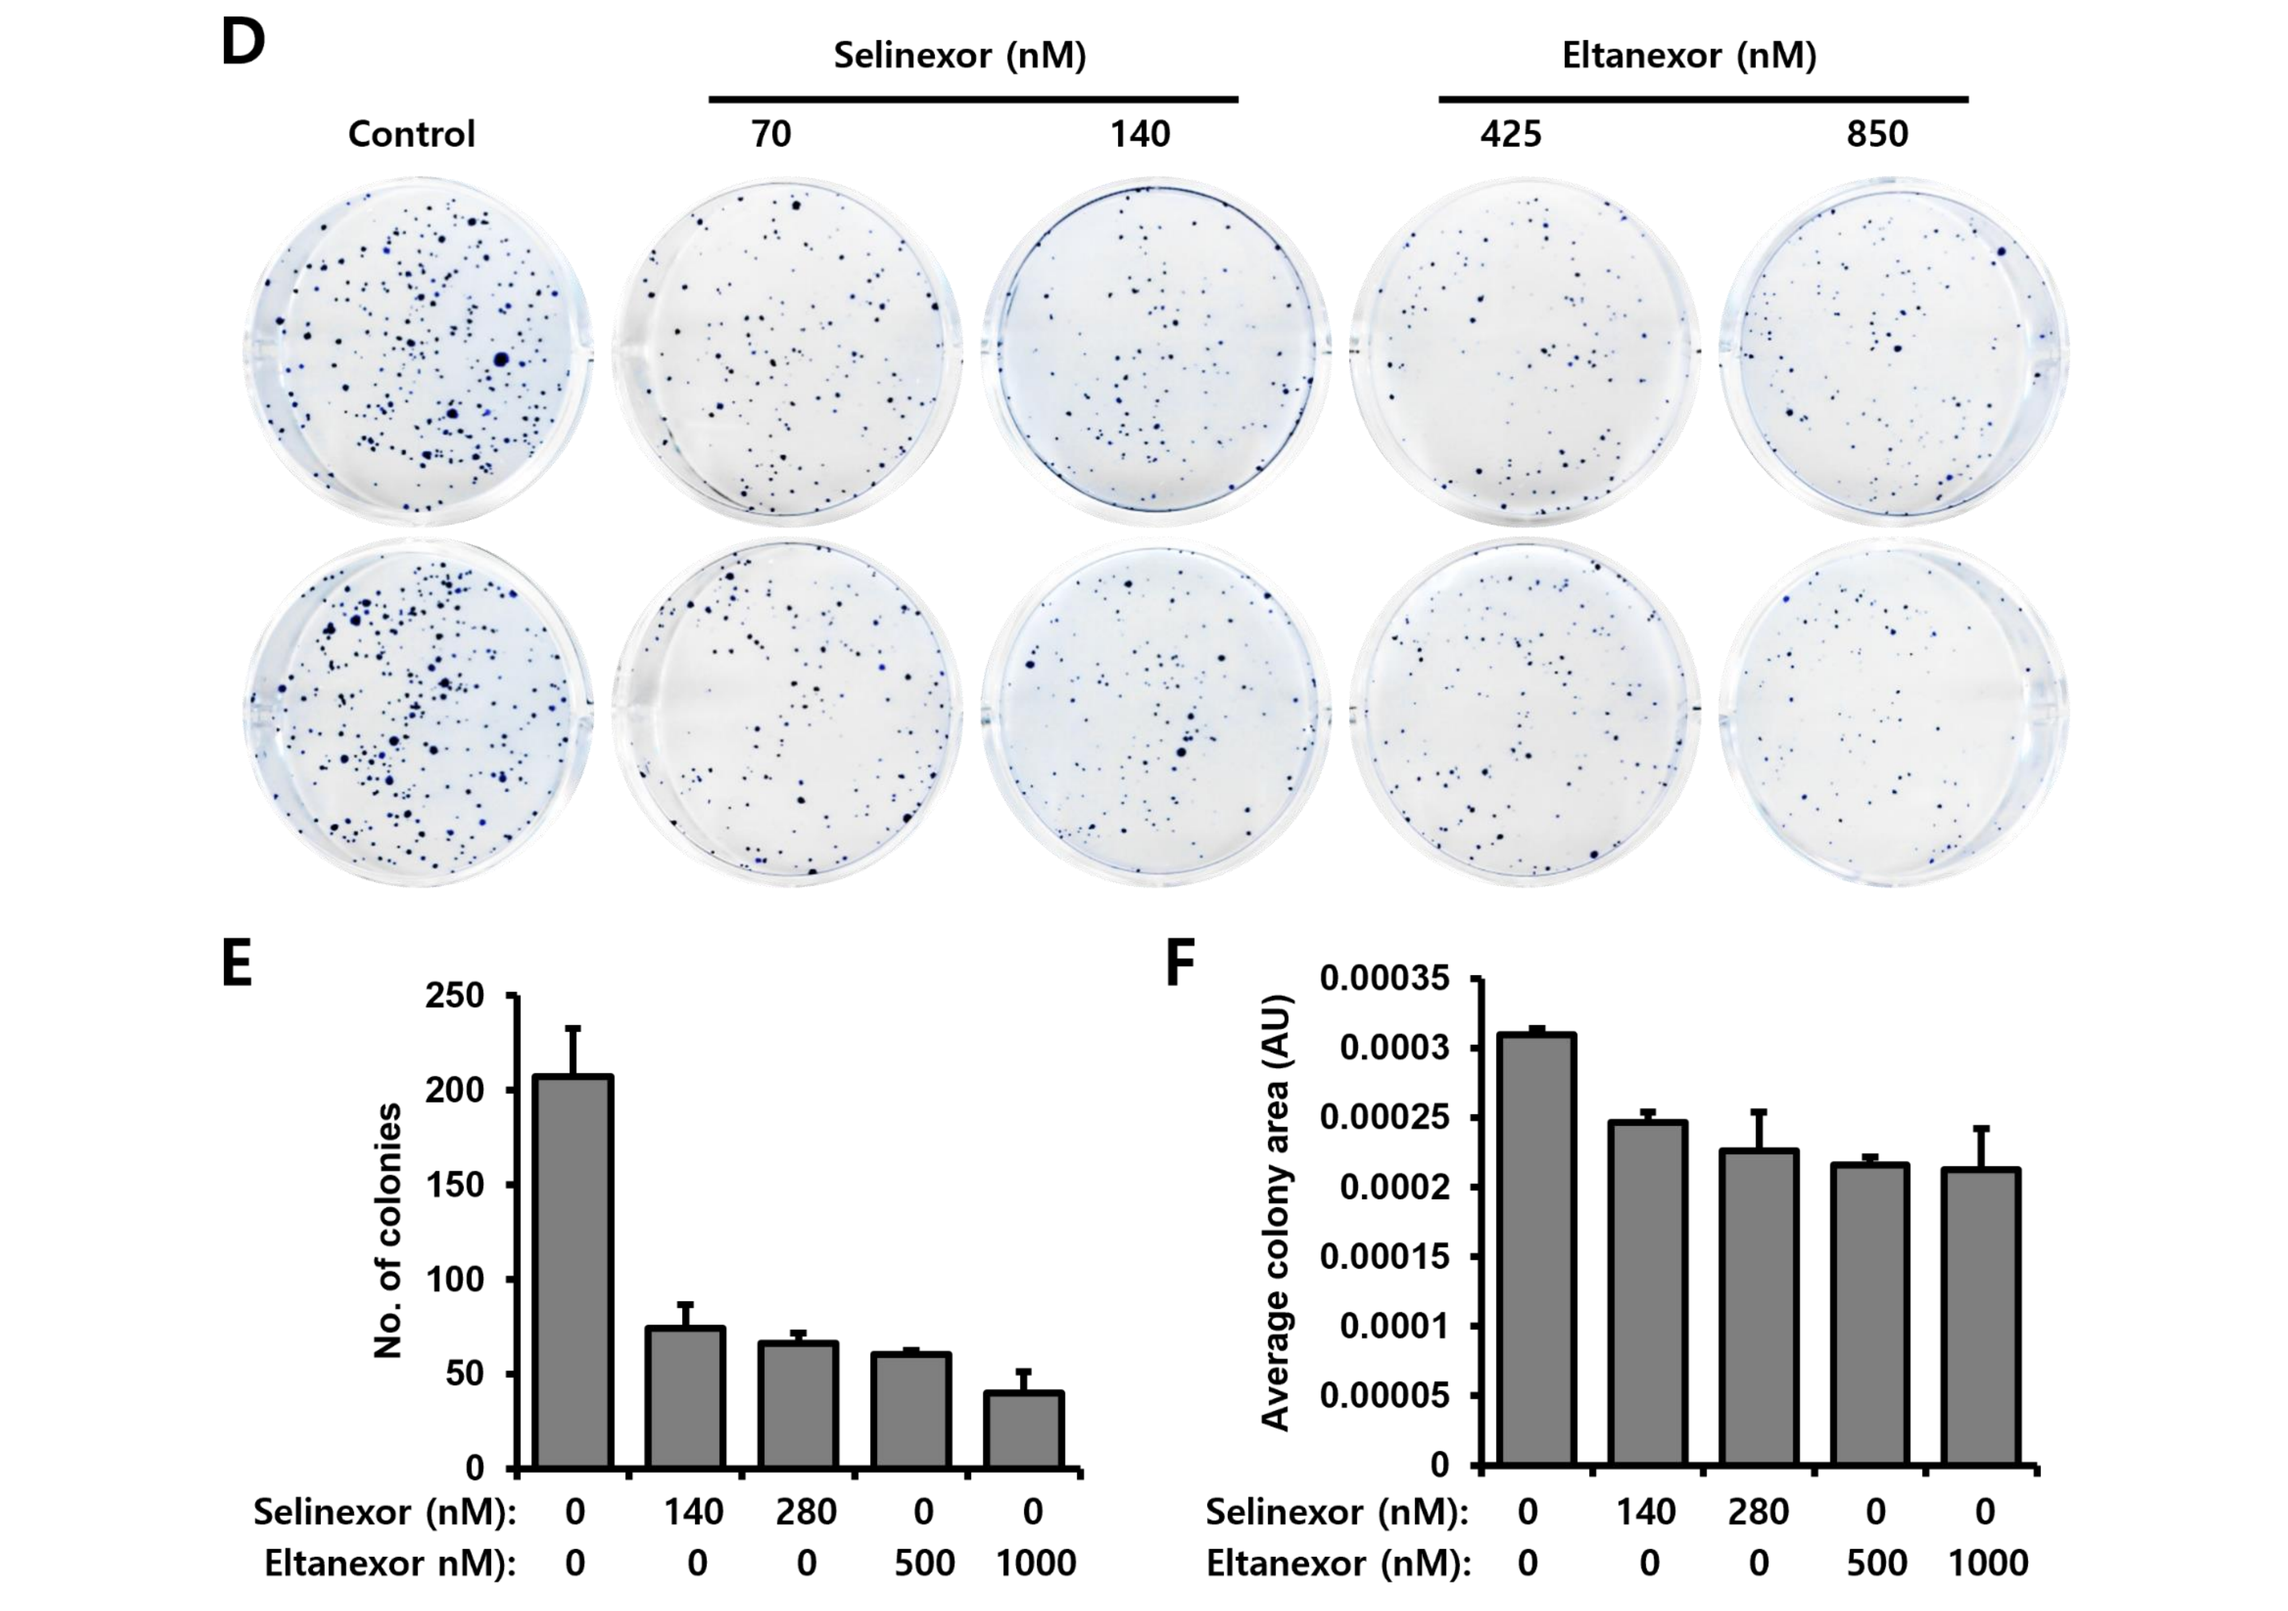
D
Selinexor (nM)
Eltanexor (nM)
Control
70
140
425
850
E
No. of colonies
F
Average colony area (AU)

Figure 3. Colony formation ability in BON1 and QGP1 cells under KPT-330 and KPT-8602 treatment. A,D: Representative colonies. B,E: Bar diagram showing the number of colonies with indicated doses of drugs. C-F. Bar diagram showing the average area of the colonies with indicated doses of drugs.

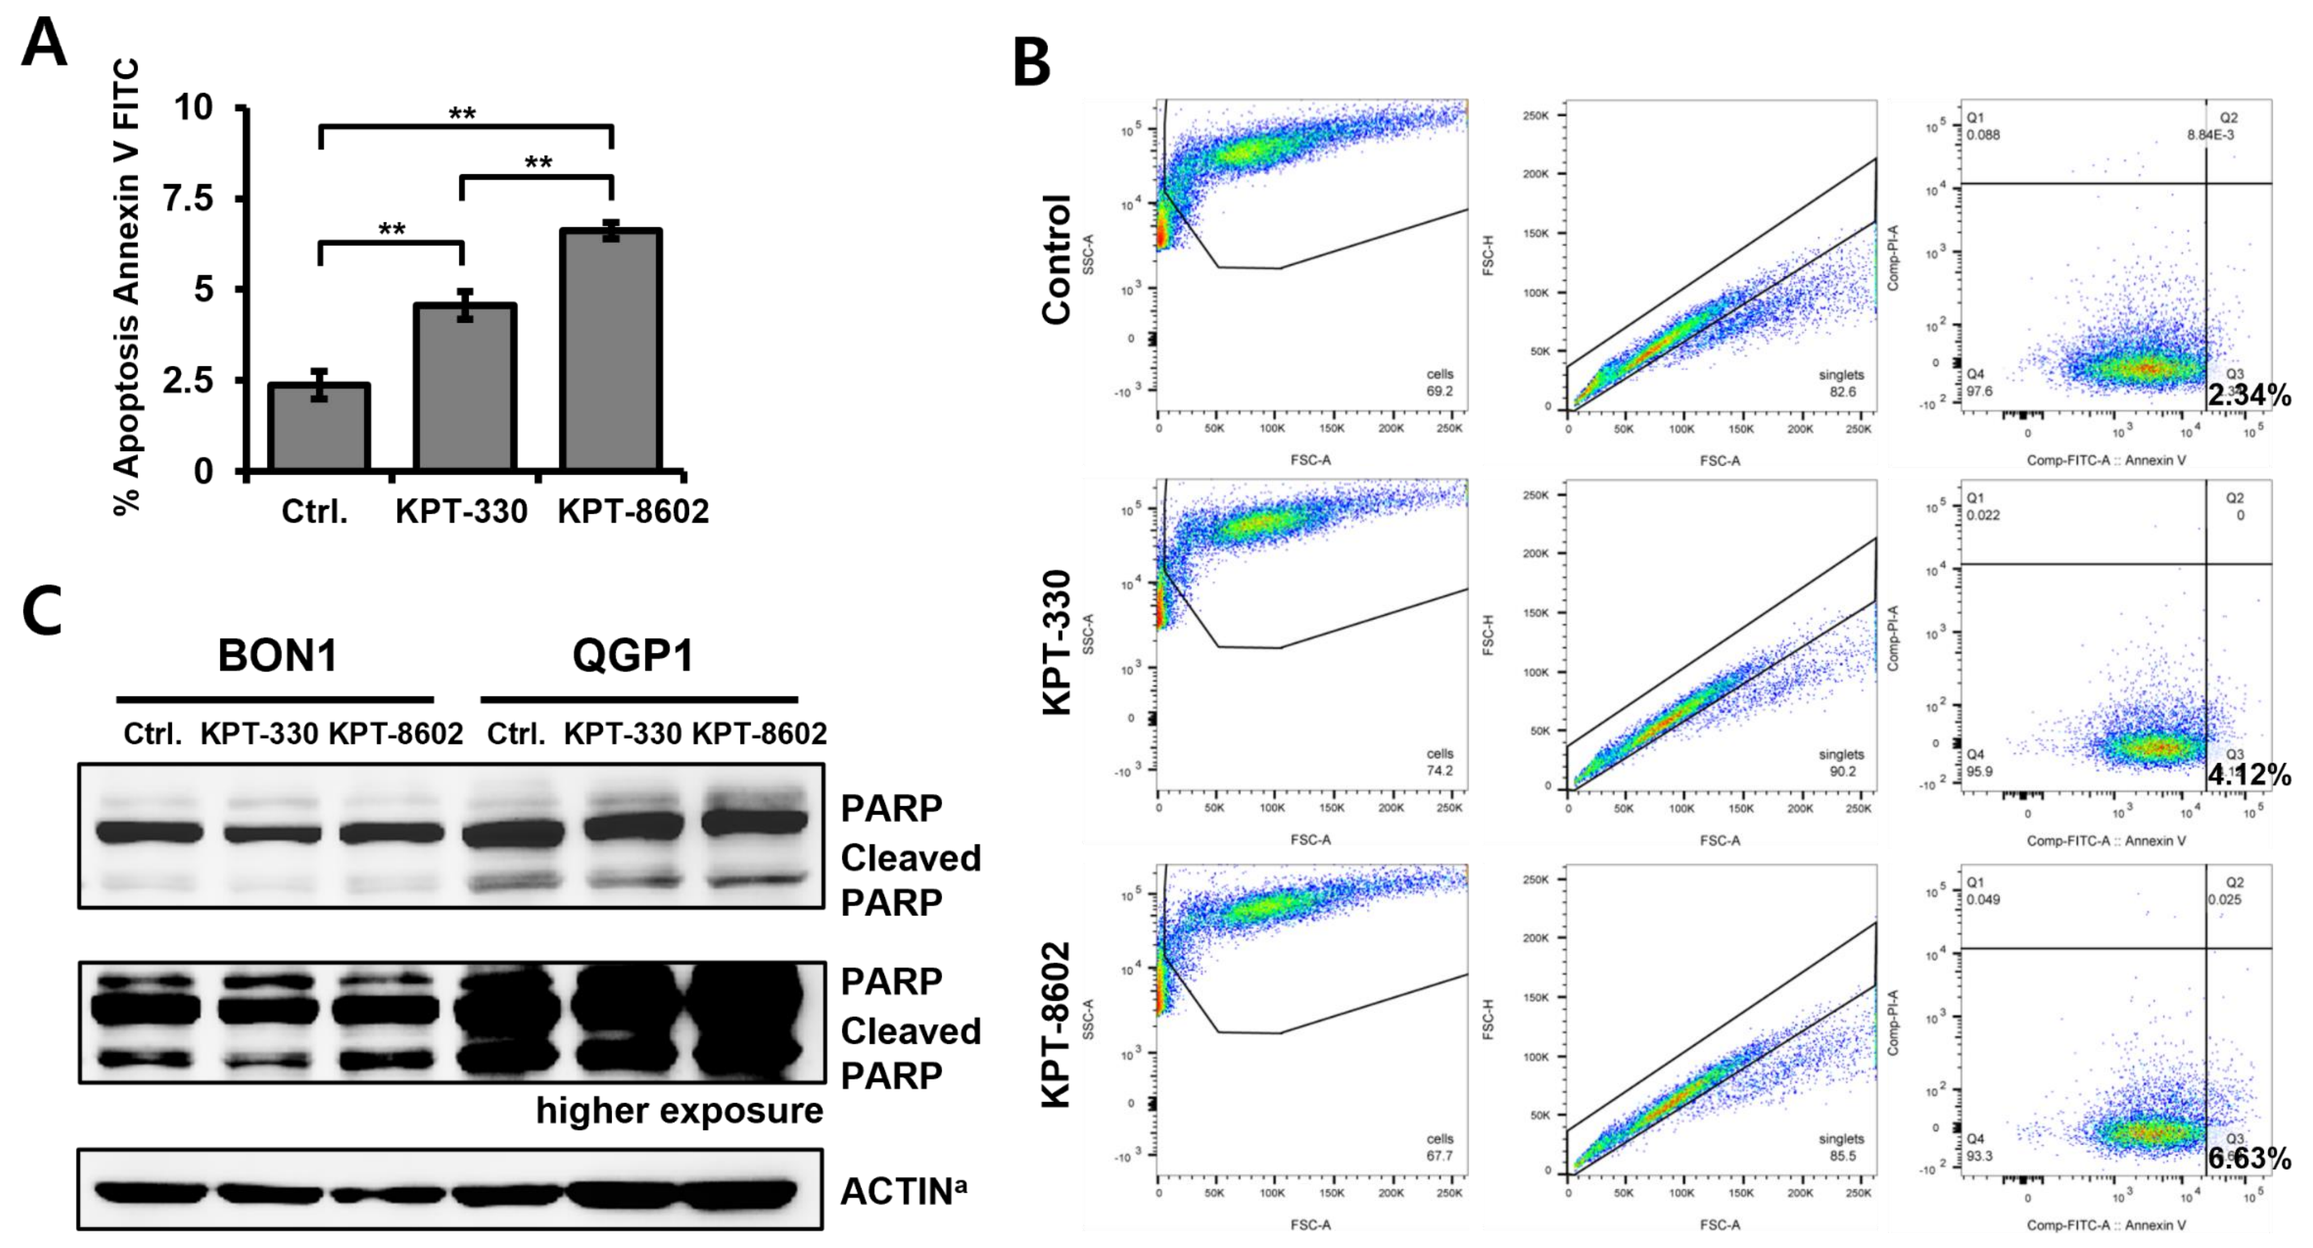
A
% Apoptosis Annexin V FITC
Ctrl.
KPT-330
KPT-8602
B
Control
KPT-330
KPT-8602
2.34%
4.12%
6.63%
C
BON1
QGP1
PARP
Cleaved PARP
higher exposure
ACTINa

Figure 4. Apoptotic cell deaths in PNET cells under KPT-330 and KPT-8602 treatment. A. Bar diagram showing the apoptotic cell deaths in BON1 cells. B. Representative FACS images. C. Western blot analysis of PARP cleavage in BON1 and QGP1 cells. Cells were treated with KPT-330 and KPT-8602 with a concentration of 280 nM and 1000 nM respectively for 72 hours.

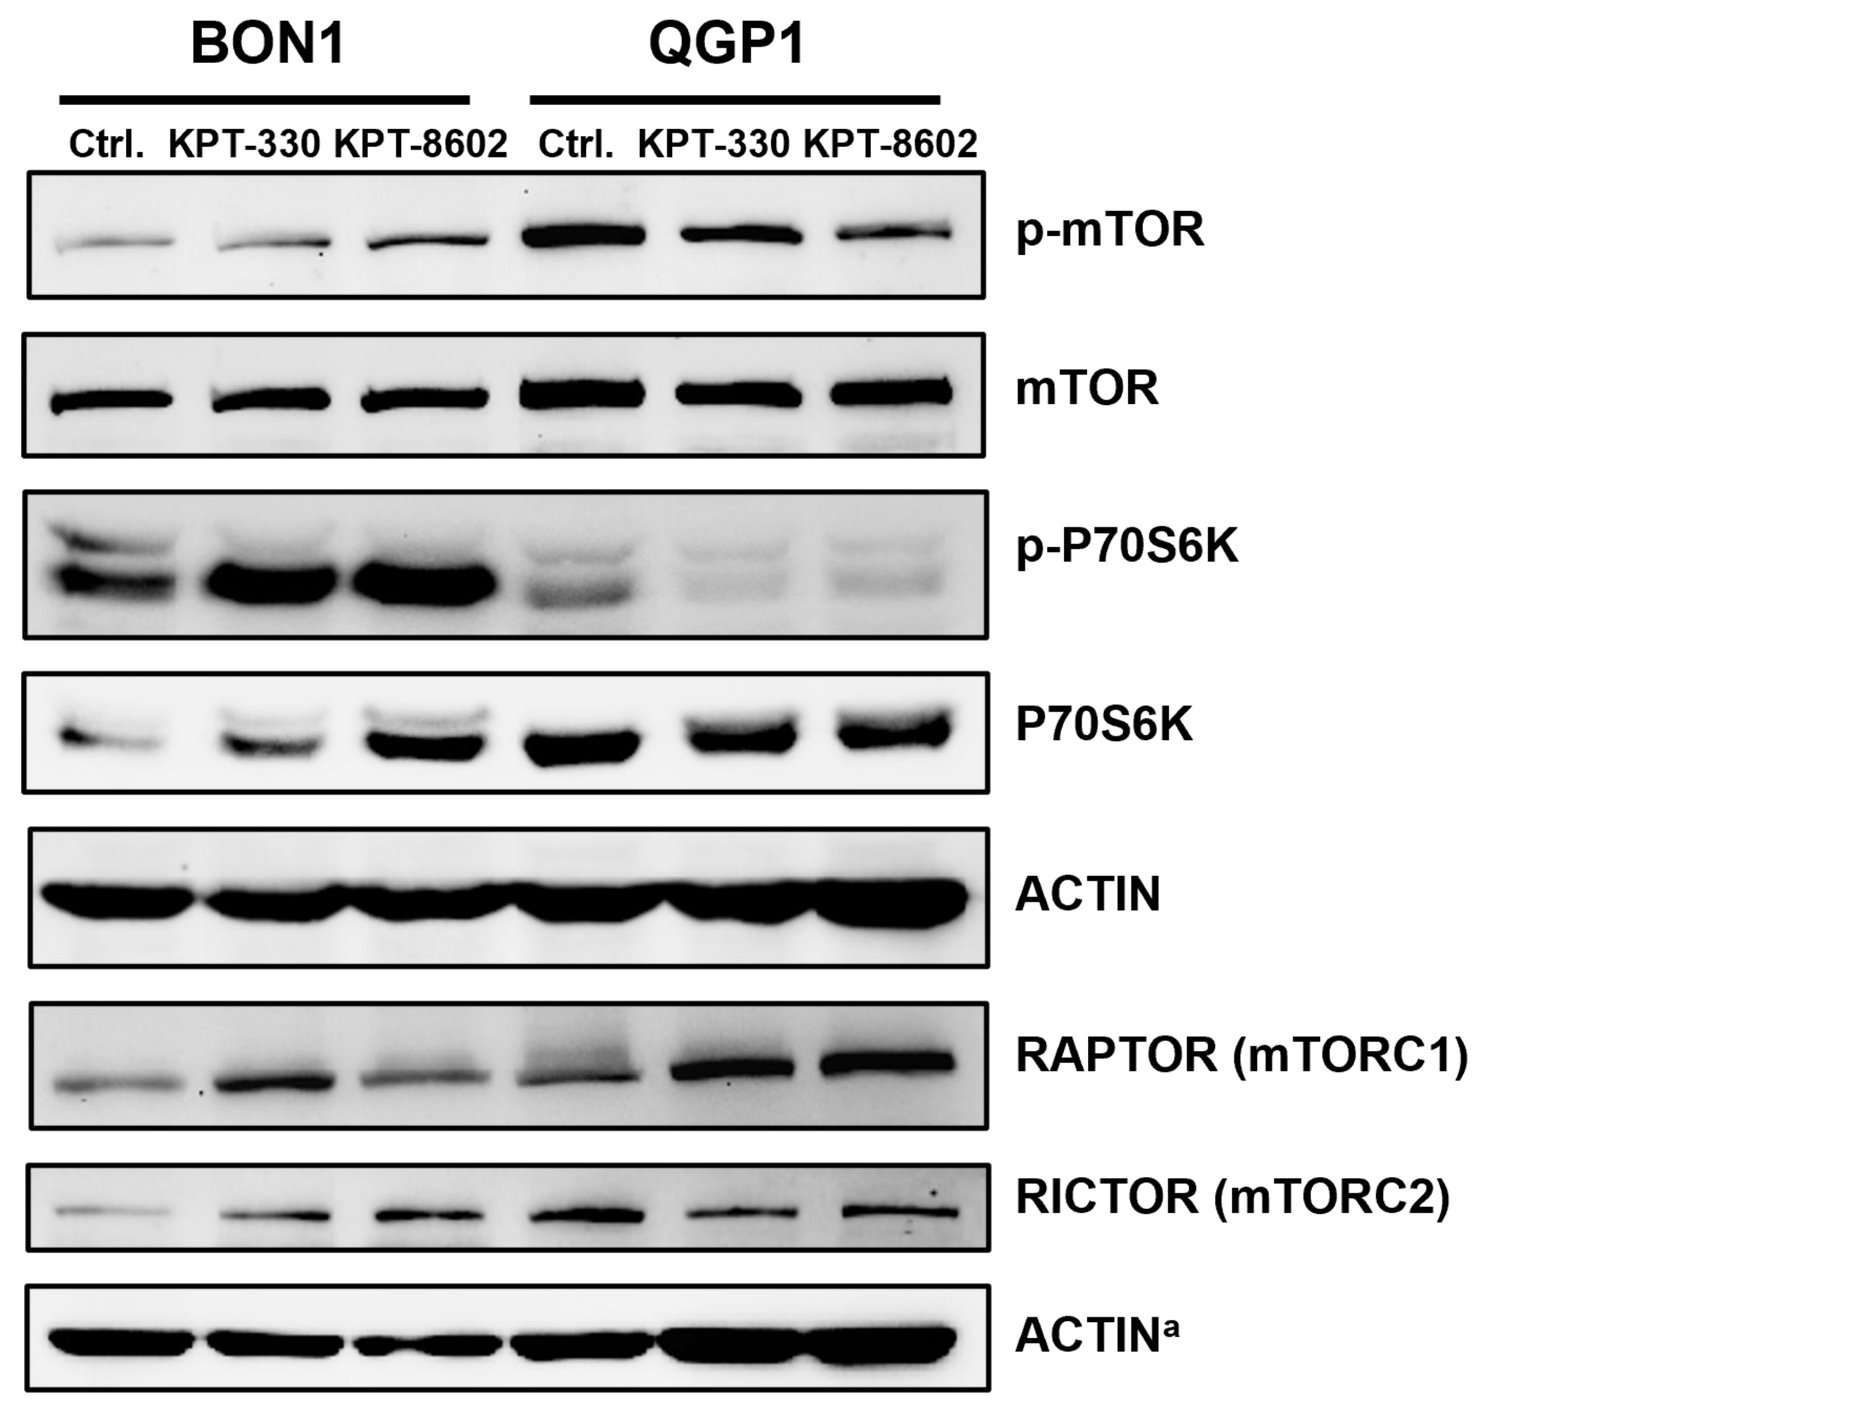
BON1
QGP1
Ctrl. KPT-330 KPT-8602 Ctrl. KPT-330 KPT-8602
p-mTOR
mTOR
p-P70S6K
P70S6K
ACTIN
RAPTOR (mTORC1)
RICTOR (mTORC2)
ACTINa

Figure 5. Western blot analysis of mTOR pathway associated proteins in BON1 and QGP1 cells treated with KPT-330 and KPT-8602. Cells were treated with KPT-330 and KPT-8602 with a concentration of 280 nM and 1000 nM respectively for 72 hours. Cell lysis was done in RIPA buffer with inhibitors.

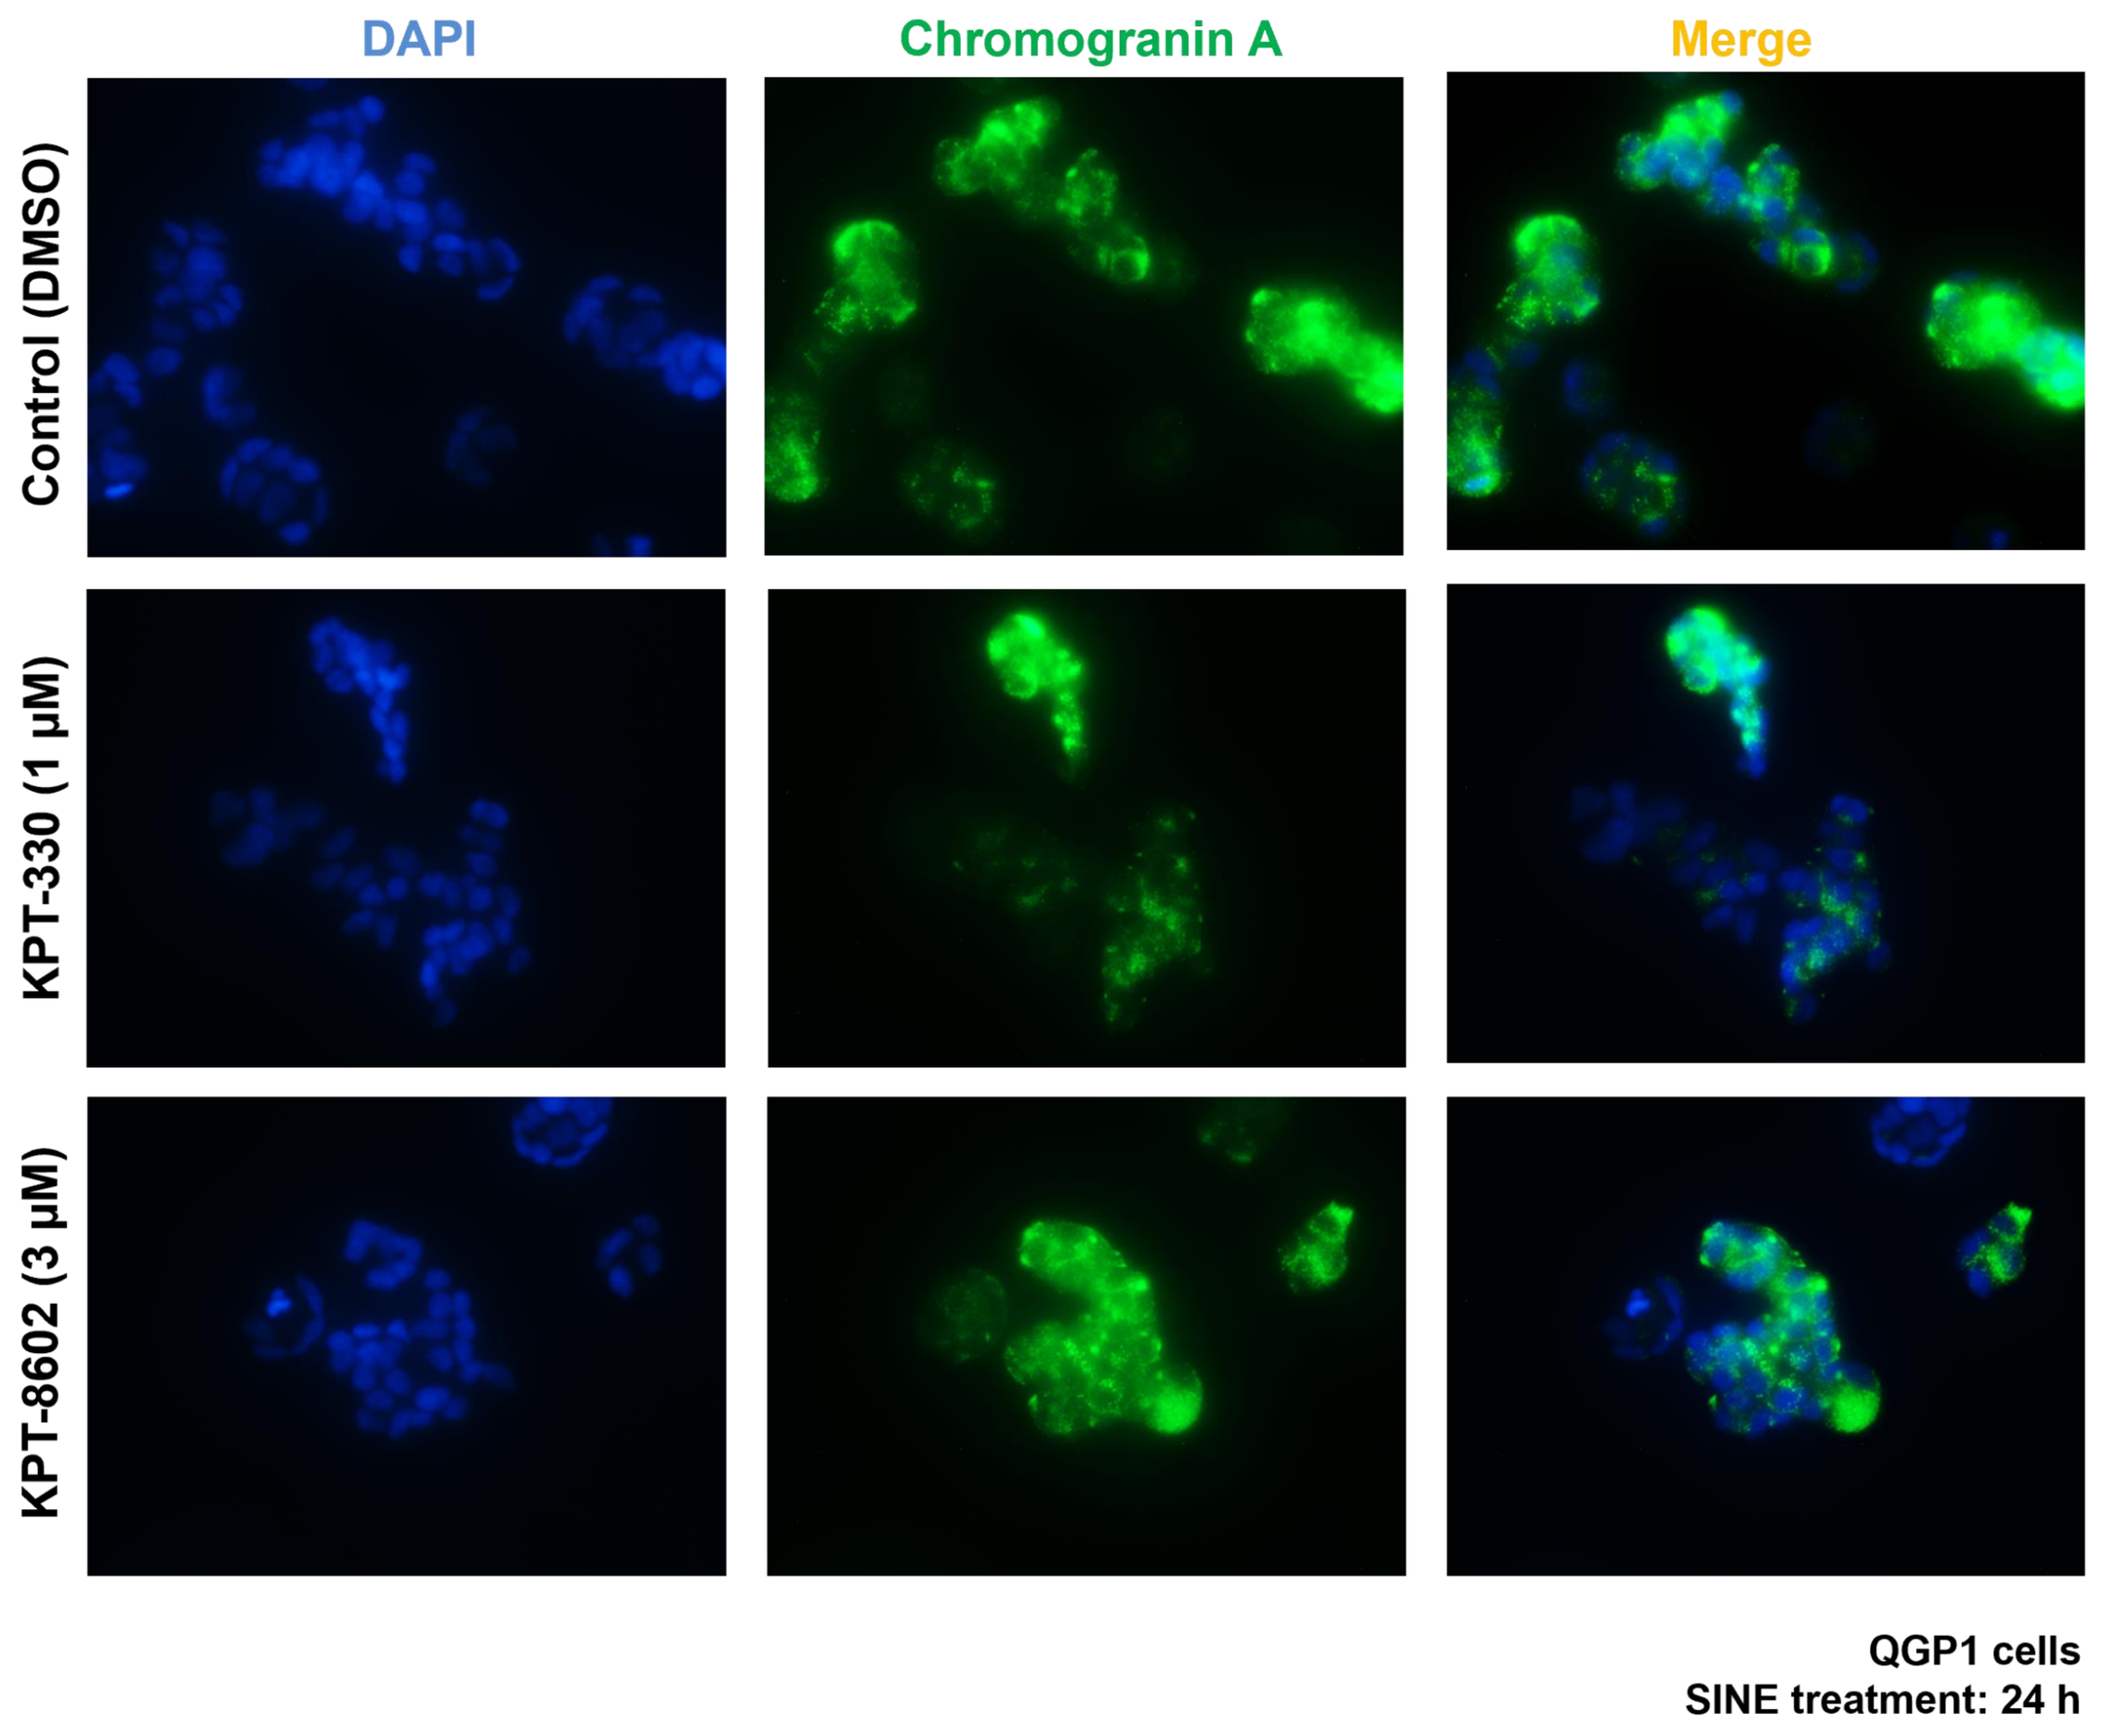
DAPI
Chromogranin A
Merge
Control (DMSO)
KPT-330 (1 µM)
KPT-8602 (3 µM)
QGP1 cells
SINE treatment: 24 h

Figure 6. Immunofluorescence (IF) detection of Chromogranin A in QGP1 cells treated with KPT-330 and KPT-8602. Cells were grown in poly-L-lysine coated chamber slide, treated with indicated concentration of SINE compounds for 24h. Fixation was done in 4% paraformaldehyde and stained with 1:100 dilution of Chromogranin A (green) antibody. Nucleus was stained with DAPI (blue).

Conclusion

The current study is the first approach to target pancreatic neuroendocrine tumors (PNETs) using nuclear export inhibitors also known as selective inhibitor of nuclear export (SINE). Both the 1st and 2nd generation XPO1 inhibitors namely KPT-330 (selinexor/XPOVIO), KPT-8602 (eltanexor) were able to suppress the growth and induction of apoptosis in BON1 and QGP1 PNET tumor cells. These *in vitro* findings warrant further pre-clinical and clinical investigations to determine the effectiveness of SINE compounds in PNETs.

References

1. https://www.cancer.gov/types/pancreatic/hp/pnet-treatment-pdq
2. Mpilla GB, Philip PA, El-Rayes B, Azmi AS. 2020. Pancreatic neuroendocrine tumors: Therapeutic challenges and research limitations. World J Gastroenterol. 28;26(28):4036-4054. doi: 10.3748/wjg.v26.i28.4036. PMID: 32821069.
3. Azmi AS, Uddin MH, Mohammad RM. 2021. Nuclear Export Protein XPO1- from Biology to Targeted Therapy. Nat Rev Clin Oncol. Mar;18(3):152-169. doi: 10.1038/s41571-020-00442-4. PMID: 33173198 (review).
4. GB Mpilla, Uddin MH, Al-Hallak MN, Aboukameel A, Li Y, … Azmi AS. 2021. PAK4-NAMPT Dual Inhibition Sensitizes Pancreatic Neuroendocrine Tumors to Everolimus. Mol Cancer Ther. 20(10):1836-1845. doi: 10.1158/1535-7163.

Acknowledgement

Work in the lab of Asfar S. Azmi is supported by R01CA24060701, NIH MERIT Award (R37 5R37CA215427), Partners Funds and SKY Foundation Inc.

RESEARCH POSTER PRESENTATION DESIGN © 2019
www.PosterPresentations.com