Cabozantinib Plus Telgaglenstat (CB-839): A Phase II Combination in Patients with Well-Differentiated Gastroenteropancreatic Neuroendocrine Tumors (WD-GEP NETs)

Satya Das (satya.das@vumc.org)[1], Jennifer Whisenant[1], Dan Ayers[2], H. Charles Manning[3] and Jordan Berlin[1]

[1]Vanderbilt University Medical Center, Department of Internal Medicine, Division of Hematology and Oncology
[2]Vanderbilt University Medical Center, Department of Biostatistics [3] The University of Texas MD Anderson Cancer Center, Division of Cancer Systems Imaging

SUMMARY

- GEP NET tumors are driven by vascular endothelial growth factor (VEGF) mediated oncogenesis. Tumors which rely on VEGF rely on higher levels of glutamine metabolism to support their metabolic needs.
- Glutamine metabolism in NET cells, along with other tumors such as renal cell carcinoma (RCC) and hepatocellular carcinoma (HCC), is mediated by the enzyme glutaminase which converts glutamine to glutamate.
- The glutaminase inhibitor CB-839 has demonstrated in vivo and early clinical synergy in combination with cabozantinib in patients with RCC.
- Given the single agent cytoreductive potential demonstrated by cabozantinib in patients with WD-GEP NETs, we have proposed a phase II single-arm study of cabozantinib plus CB-839 in this patient population.
- The study includes 37 patients and utilizes a Simon's two-stage design with a primary endpoint of objective response rate (ORR). Beyond efficacy, we aim to identify biomarkers (tissue, and possibly imaging-based) which may identify the patients with tumors that are particularly responsive to the study combination

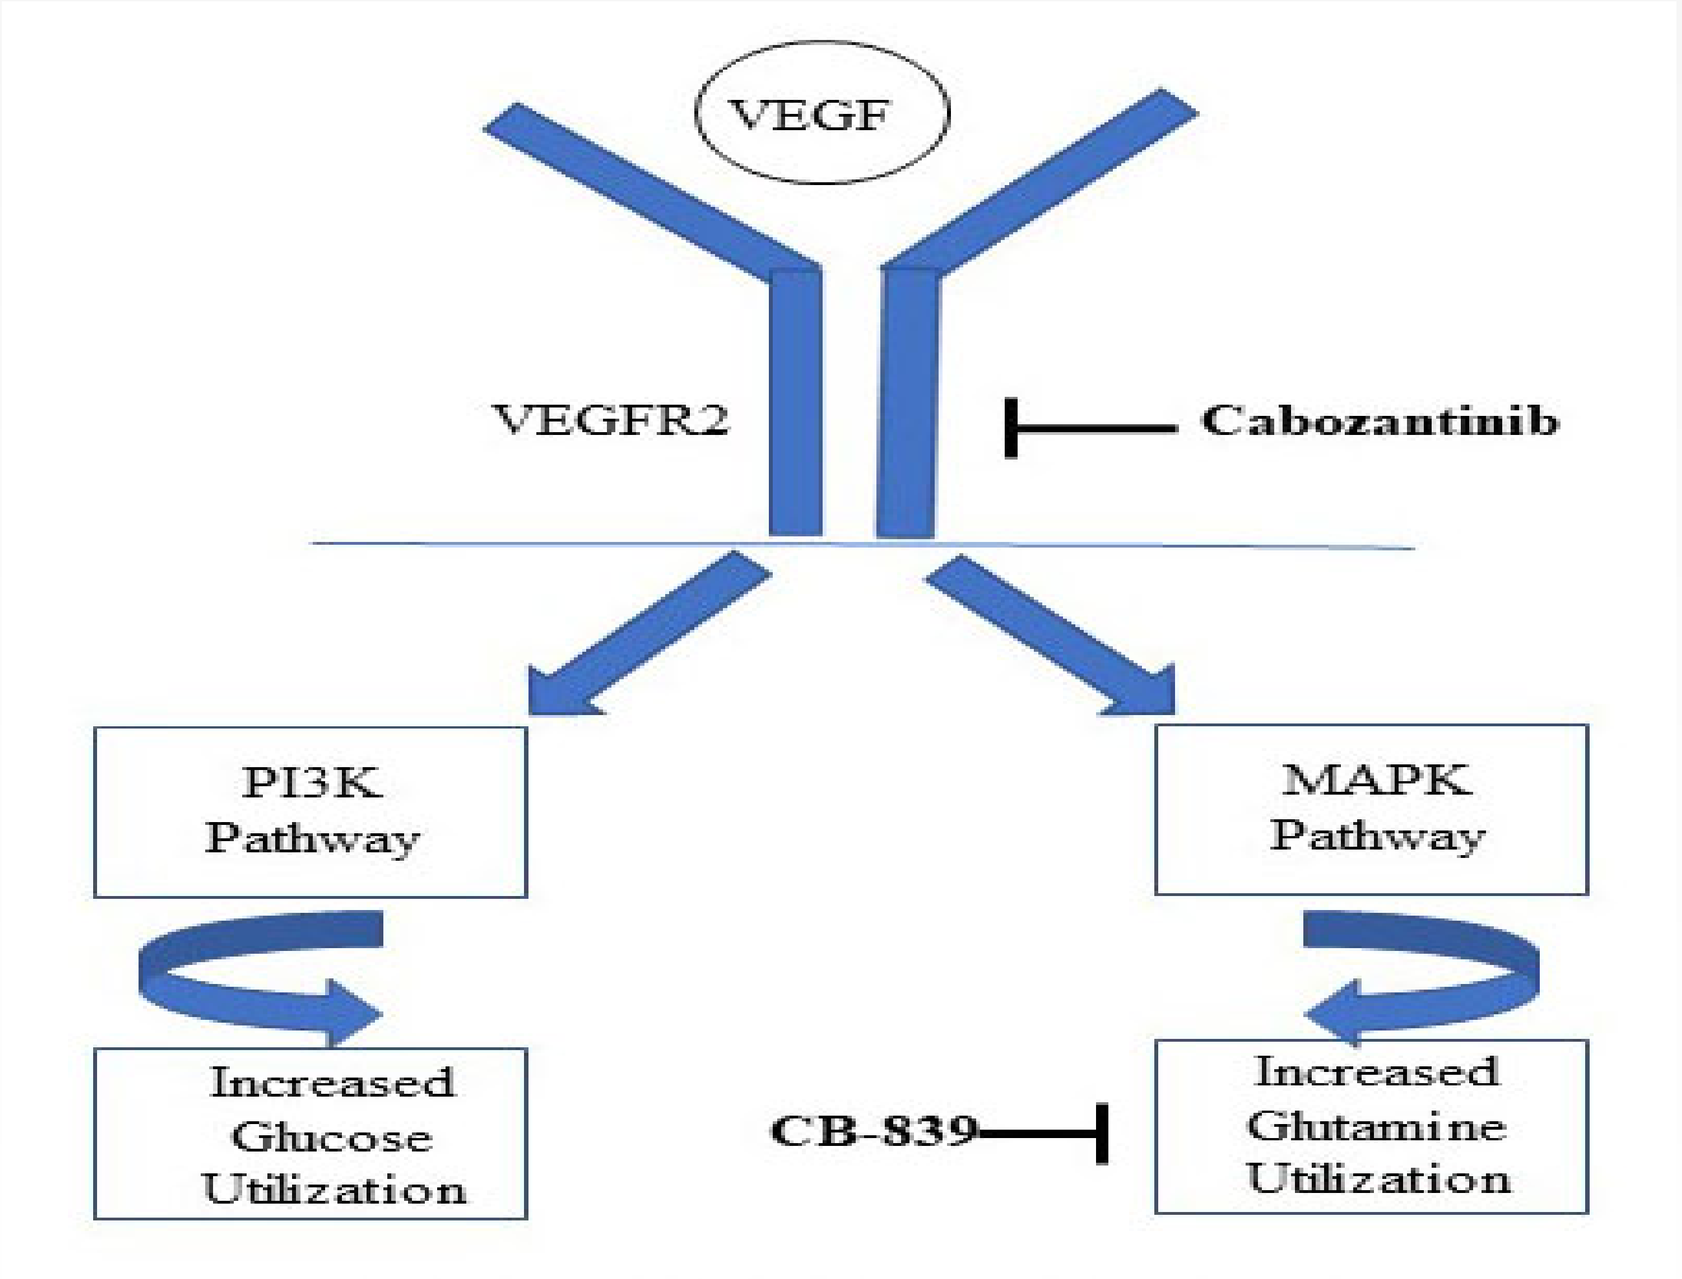
VEGF
VEGFR2
Cabozantinib
PI3K Pathway
MAPK Pathway
Increased Glucose Utilization
CB-839
Increased Glutamine Utilization

Figure 1. Rationale behind the study combination. VEGF and other growth factor driven tumors depend on glutamine utilization to support cell growth and proliferation.
Abbreviations: PI3K, PI3 kinase; MAPK, MAP kinase

RESEARCH POSTER PRESENTATION DESIGN © 2019
www.PosterPresentations.com

BIOLOGIC RATIONALE

- Patients with WD-GEP NETs have limited cytoreductive options beyond capecitabine and temozolomide & peptide radionuclide receptor therapy (PRRT) [1].
- Cabozantinib demonstrated promising cytoreductive, as well as progression-prolonging capacity, in WD GEP-NET patients in a phase II study reported by Chan et al [2].
- One of the primary pathways of oncogenesis in GEP-NET tumors is mediated by the VEGF receptor. Tumors that signal through VEGF and other receptor tyrosine kinase pathways tend to utilize higher levels of glutamine metabolism to support enhanced cell growth and proliferation [3,4] **(Figure 1).**
- Glutamine utilization in tumor cells is mediated in large part by the enzyme glutaminase, which converts glutamine to glutamate; glutaminase is over-expressed in GEP-NETs and other malignancies such as RCC and HCC [5,6].
- Glutamate has pro-tumorigenic effects in NET cell lines and in vivo models, and blocking its primary receptors with antagonists, inhibits cell proliferation and invasiveness, increases apoptosis and reduces tumor volume [7,8]. The glutaminase inhibitor CB-839 has demonstrated synergy with cabozantinib in cell line and xenograft models of RCC, as well as early clinical efficacy in RCC patient cohorts [9,10,11] **(Figure 2).**
- If the study is positive, the combination would represent a new cytoreductive option for all GEP NET patients.

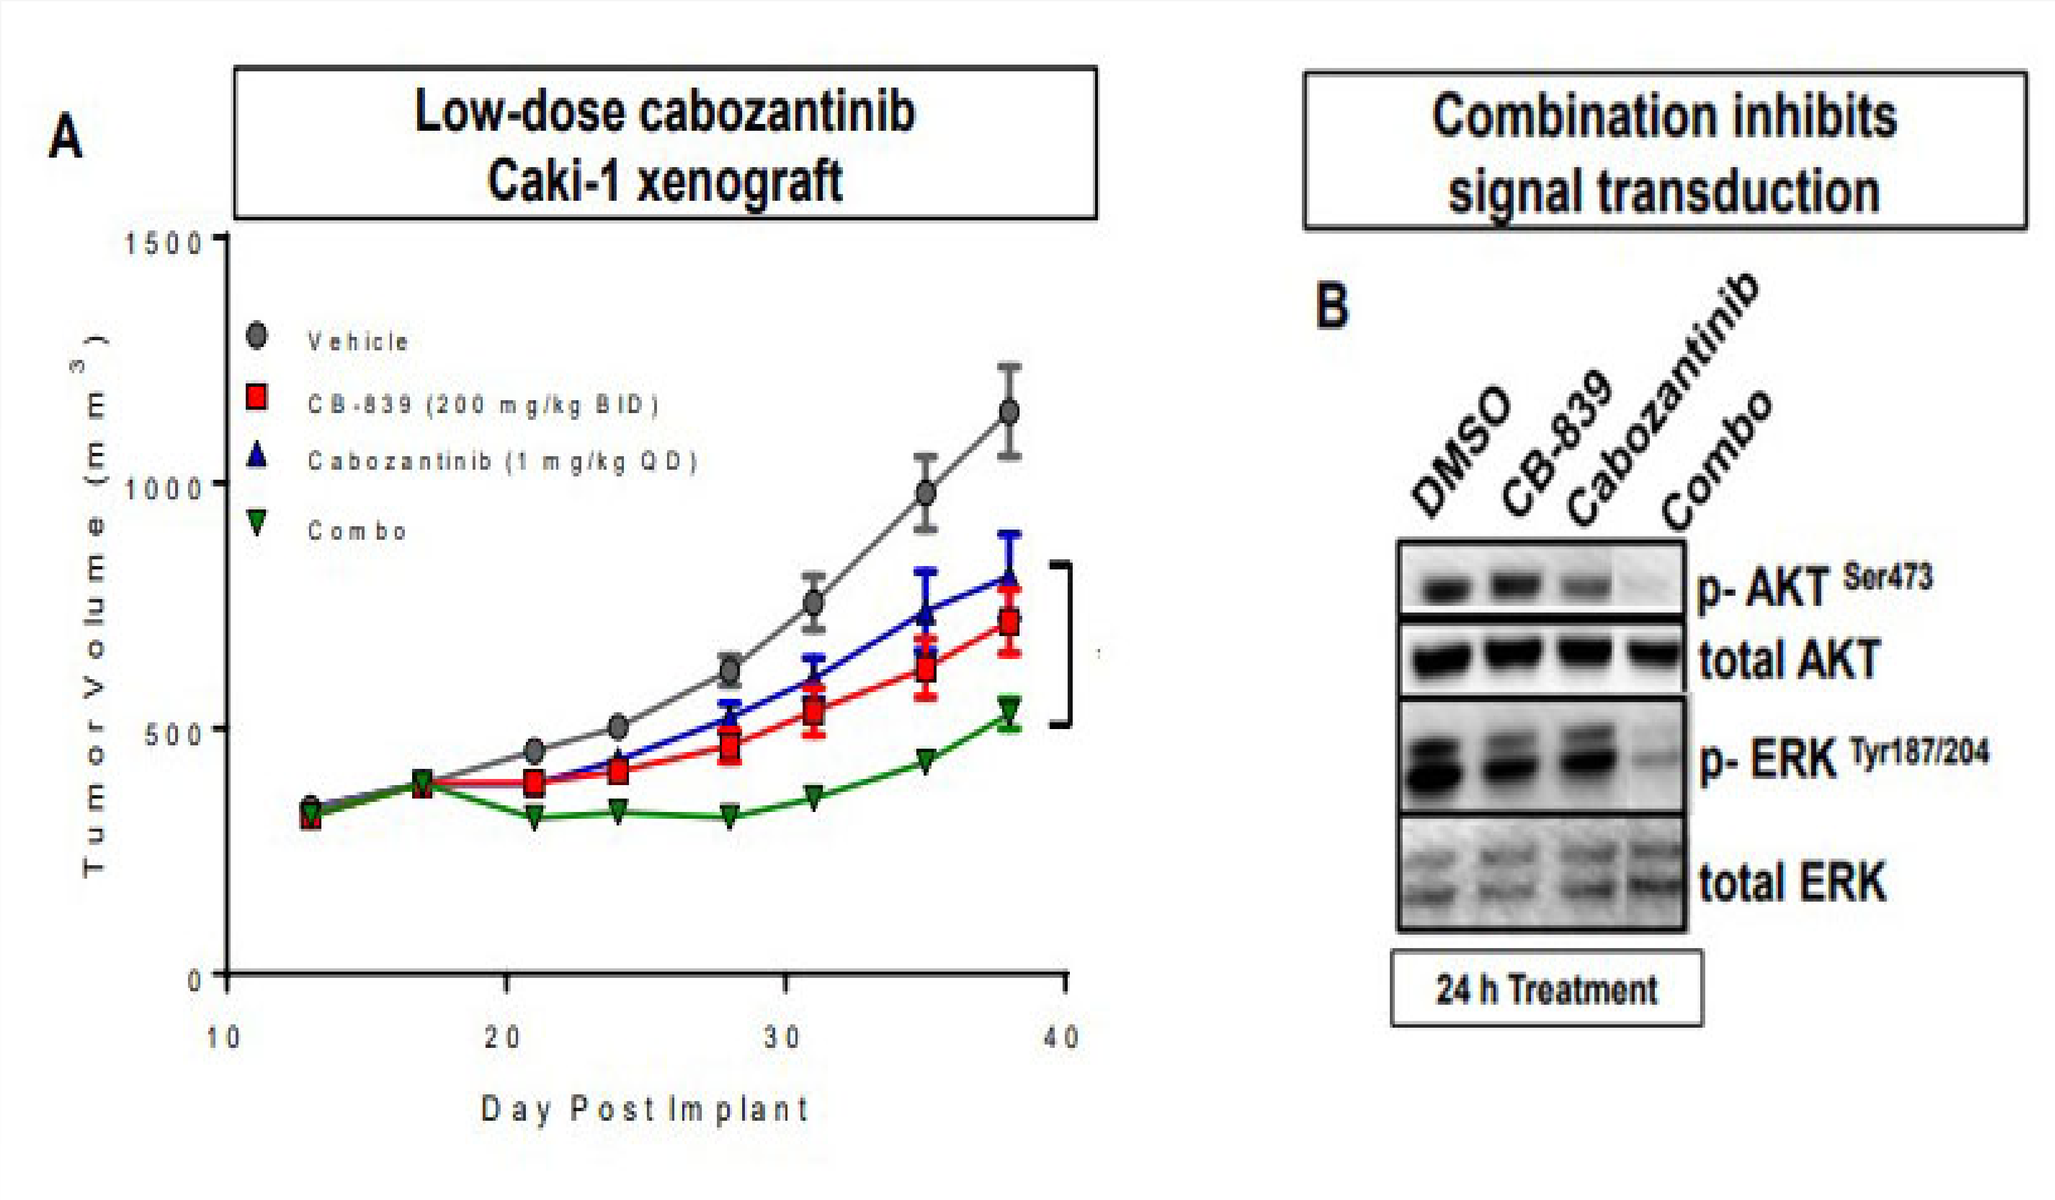
A
Low-dose cabozantinib
Caki-1 xenograft
Vehicle
CB-839 (200 mg/kg BID)
Cabozantinib (1 mg/kg QD)
Combo
Tumor Volume (mm3)
Day Post Implant
Combination inhibits signal transduction
B
DMSO
CB-839
Cabozantinib
Combo
p- AKT Ser473
total AKT
p- ERK Tyr187/204
total ERK
24 h Treatment

Figure 2. Data from two separate experiments in Caki-1 RCC xenografts (A) and cell lines (B). Panel A suggests the combination of CB-839 and low dose cabozantinib slows tumor growth beyond the tumor growth reduction with either agent alone. Panel B demonstrates the ability of ability of the combination to efface signal transduction in PI3K and MAPK pathways.
Abbreviations: RCC, renal cell carcinoma; PI3K, PI3 kinase; MAPK, MAP kinase

OBJECTIVES

Primary
- Assess the ORR of study patients on 16-week restaging scans by RECIST 1.1

Secondary
- Assess progression-free survival and overall survival of study patients, by primary tumor site (PNET and non-PNET)
- Establish a recommended phase II dose (RP2D) during the safety lead-in period
- Characterize grade (G)3/4 adverse events (AEs) or G2 AEs lasting ≥ 7 days in study patients (AEs will be graded by CTCAE v5.0)

Exploratory
- Assess glutamine metabolism gene signatures through RNA-Seq from pretreatment tumor biopsies in 7 stage I patients

METHODS

Key Inclusion Criteria
- Any patients with WD-GEP NETs (including tumors with Ki-67 >20%) with biopsy proven disease.
- Patients must have unresectable or metastatic disease.
- Patients must have received and progressed on at least one prior line of systemic therapy (PRRT does not count towards a prior therapy line).
- Patients must have performance status (PS) of 0-1.
- Patients with adequate organ function such as hematologic reserve (Hgb > 9, platelets > 150,000), renal function (eGFR > 40) and hepatic function (bilirubin < 2).

Study Treatment
- Patients will receive cabozantinib 60 mg daily and CB-839 at a starting dose of 800 mg BID daily on 28-day cycles.
- Although this combination has phase I safety data in RCC, it does not in GEP-NETs. As such, a safety lead-in has been built into stage I of our study to establish a RP2D.

Dose Level	Cabozantinib (PO daily)	CB-839 (PO BID)
1	60 mg	800 mg
-1	60 mg	600 mg
-2	40 mg	600 mg

METHODS CONT.

- Patients will be enrolled in cohorts of 3 during this lead-in phase. At least 6 patients must be treated at a specific dose level with a dose-limiting toxicity (DLT) rate < 33% for that dose to be deemed the RP2D.
- Outcomes in patients treated within the safety lead-in period will be included for the efficacy analysis.
- A DLT for patients in this study is defined as any G4 hematologic toxicity, with the exception of lymphopenia, or any G3 non-hematologic toxicity, with the exception of diarrhea ≤ 72 hours, nausea ≤ 72 hours, fatigue ≤ 72 hours, hypertension ≤ 72 hours or transaminase elevations ≤ 7 days.

Study Schema

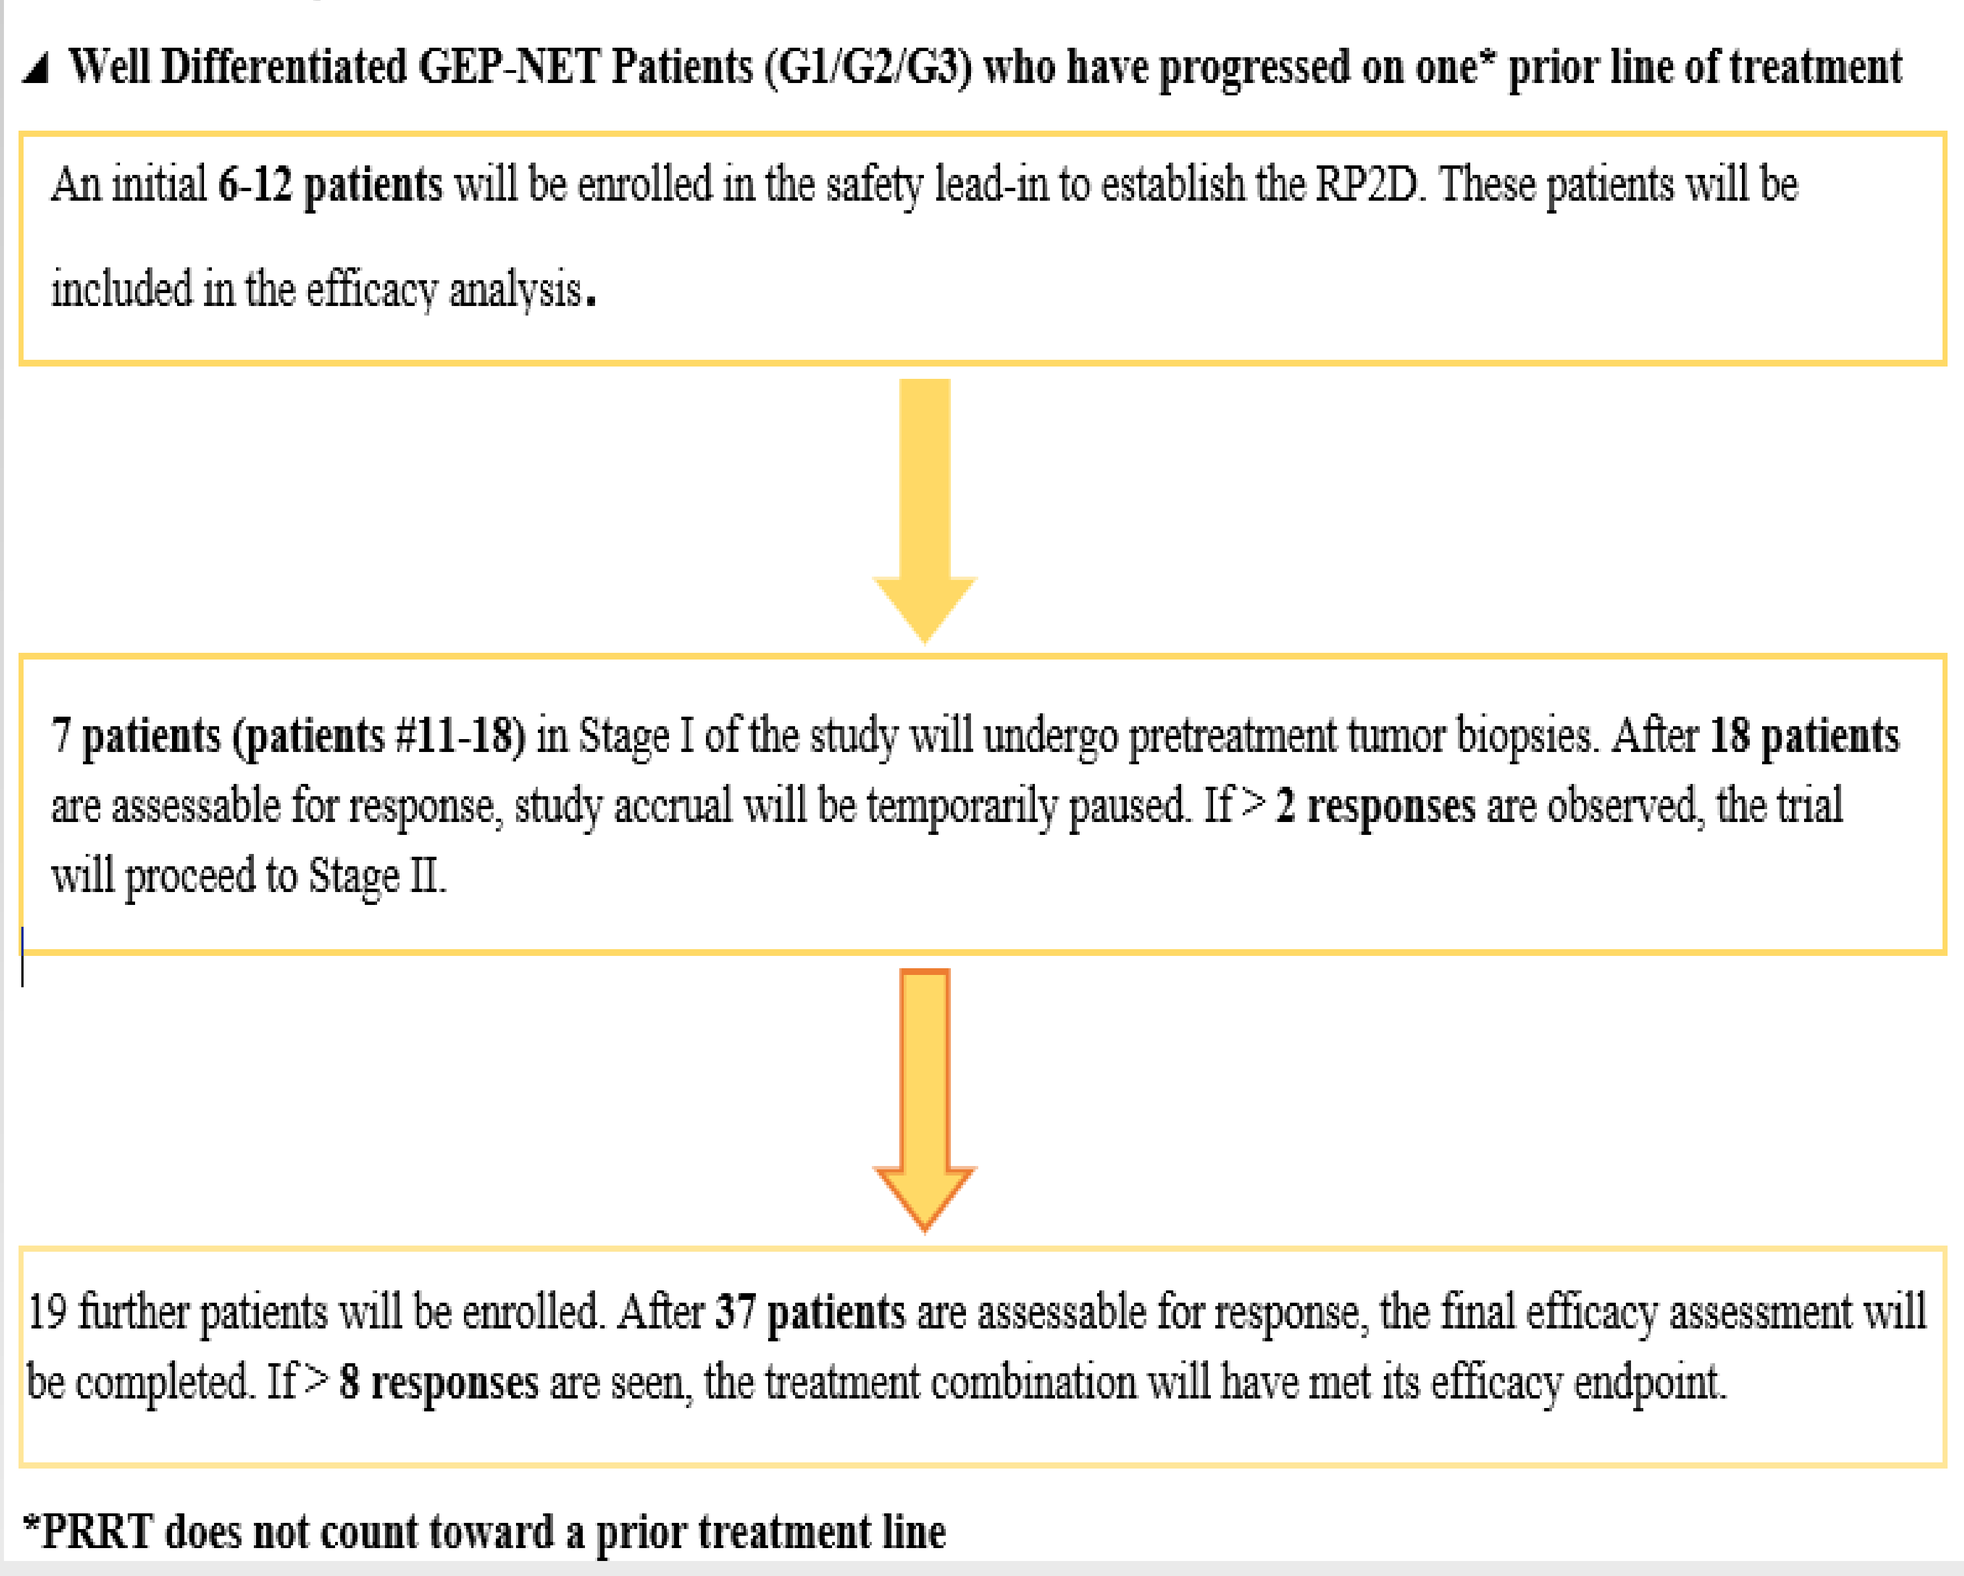
Well Differentiated GEP-NET Patients (G1/G2/G3) who have progressed on one* prior line of treatment
An initial 6-12 patients will be enrolled in the safety lead-in to establish the RP2D. These patients will be included in the efficacy analysis.
7 patients (patients #11-18) in Stage I of the study will undergo pretreatment tumor biopsies. After 18 patients are assessable for response, study accrual will be temporarily paused. If > 2 responses are observed, the trial will proceed to Stage II.
19 further patients will be enrolled. After 37 patients are assessable for response, the final efficacy assessment will be completed. If > 8 responses are seen, the treatment combination will have met its efficacy endpoint.
*PRRT does not count toward a prior treatment line

NEXT STEPS

- We are finalizing the study protocol and will be submitting that for IRB and Scientific Review Committee (SRC) approval at Vanderbilt Ingram Cancer Center.
- We have attained study funding from Exelixis and confirmed drug provision of CB-839 from Calithera.
- We anticipate activating the study by January 2021.

REFERENCES

1. Strosberg et al. Phase 3 Trial of 177Lu-Dotatate for Midgut Neuroendocrine Tumors. N Engl J Med 2017;376:125-135.
2. Chan et al. Phase II trial of cabozantinib in patients with carcinoid and pancreatic neuroendocrine tumors (pNET). J Clin Oncol. 2017 35(4_suppl):228-228.
3. Huang et al. Angiogenesis revisited from a metabolic perspective: role and therapeutic implications of endothelial cell metabolism. EMBO J. 2017;36:2334–2352.
4. Zecchin et al. How Endothelial Cells Adapt Their Metabolism to Form Vessels in Tumors. Front Immunol. 2017;8:1750..
5. Szeliga et al. Glutaminases in slowly proliferating gastroenteropancreatic neuroendocrine neoplasms/tumors (GEP-NETs): Selective overexpression of mRNA coding for the KGA isoform. Experimental and Mol Path. 2016;100(1):74-78.
6. Li et al. Targeting glutaminase 1 attenuates stemness properties in hepatocellular carcinoma by increasing reactive oxygen species and suppressing Wnt/beta-catenin pathway. EBioMedicine 2019;39:239-254.
7. Li et al. Hijacking the neuronal NMDAR signaling circuit to promote tumor growth and invasion. Cell 2013;153(1):86-100.
8. Haas et al. The Influence of Glutamate Receptors on Proliferation and Metabolic Cell Activity of Neuroendocrine Tumors. Anticancer Res. 2013;33(4):1267-1272.
9. Emberly et al. CB839, a selective glutamase inhibitor, has anti-tumor activity in renal cell carcinoma and synergizes with cabozantinib and everolimus. Presented at: Keystone Symposia, Tumor Metabolism: Mechanisms and Targets; March 5-9, 2017. Website: calithera.com/wp-content/uploads/2017/12/03.2017-Keystone- poster-Emberley-2017.pdf. Accessed 2/10/2019.
10. Tannir et al. Phase 1 study of glutaminase (GLS) inhibitor CB-839 combined with either everolimus (E) or cabozantinib (Cabo) in patients (pts) with clear cell (cc) and papillary (pap) metastatic renal cell cancer (mRCC). J Clin Oncol. 2018;36(suppl 6S):abstr 603.
11. Meric-Bernstam F et al. CB-839, a glutaminase inhibitor, in combination with cabozantinib in patients with clear cell and papillary metastatic renal cell cancer (mRCC): Results of a phase I study. J Clin Oncol. 2018;37(suppl 7S):abstr 549.